Potential predictive biomarker for response to radiotherapy and CXCL12-inhibition in glioblastoma in the phase I/II GLORIA trial.

Frank A. Giordano[1,2]*, Julian P. Layer[3,4]*, Sonia Leonardelli[4]*, Lea L. Friker[4,5], Roberta Turiello[4], Dillon Corvino[4], Thomas Zeyen[6], Christina Schaub[6], Wolf Müller[7], Elena Sperk[1,8], L. Christopher Schmeel[3], Katharina Sahm[9], Sied Kebir[10], Peter Hambsch[11], Torsten Pietsch[5], Sotirios Bisdas[12], Martin Glas[10], Clemens Seidel[11], Ulrich Herrlinger[6]* and Michael Hölzel[4]*

[1] Department of Radiation Oncology, University Medical Center Mannheim, University of Heidelberg; [2] DKFZ-Hector Cancer Institute at the University Medical Center Mannheim; [3] Department of Radiation Oncology, University Hospital Bonn, University of Bonn; [4] Institute of Experimental Oncology, University Hospital Bonn, University of Bonn; [5] Institute of Neuropathology, University Hospital Bonn, University of Bonn; [6] Division of Clinical Neuro-Oncology, Department of Neurology, University Hospital Bonn; [7] Institute of Neuropathology, University Hospital Leipzig, University of Leipzig; [8] Clinical Trials Unit, University Medical Center Mannheim, University of Heidelberg; [9] Department of Neurology, University Medical Center Mannheim, University of Heidelberg; [10] Division of Clinical Neurooncology, Department of Neurology, University Hospital Essen, University Duisburg-Essen; [11] Department of Radiation Oncology, University Hospital Leipzig, Leipzig, Germany; [12] Lysholm Department of Neuroradiology, National Hospital for Neurology and Neurosurgery, London

ASCO ANNUAL MEETING
Abstract #405

1 Study design

Inclusion criteria
- Newly-diagnosed supratentorial GBM (WHO CNS grade 4)
- MGMT promoter unmethylated
- Incomplete resection/biopsy only
- ECOG ≤ 2

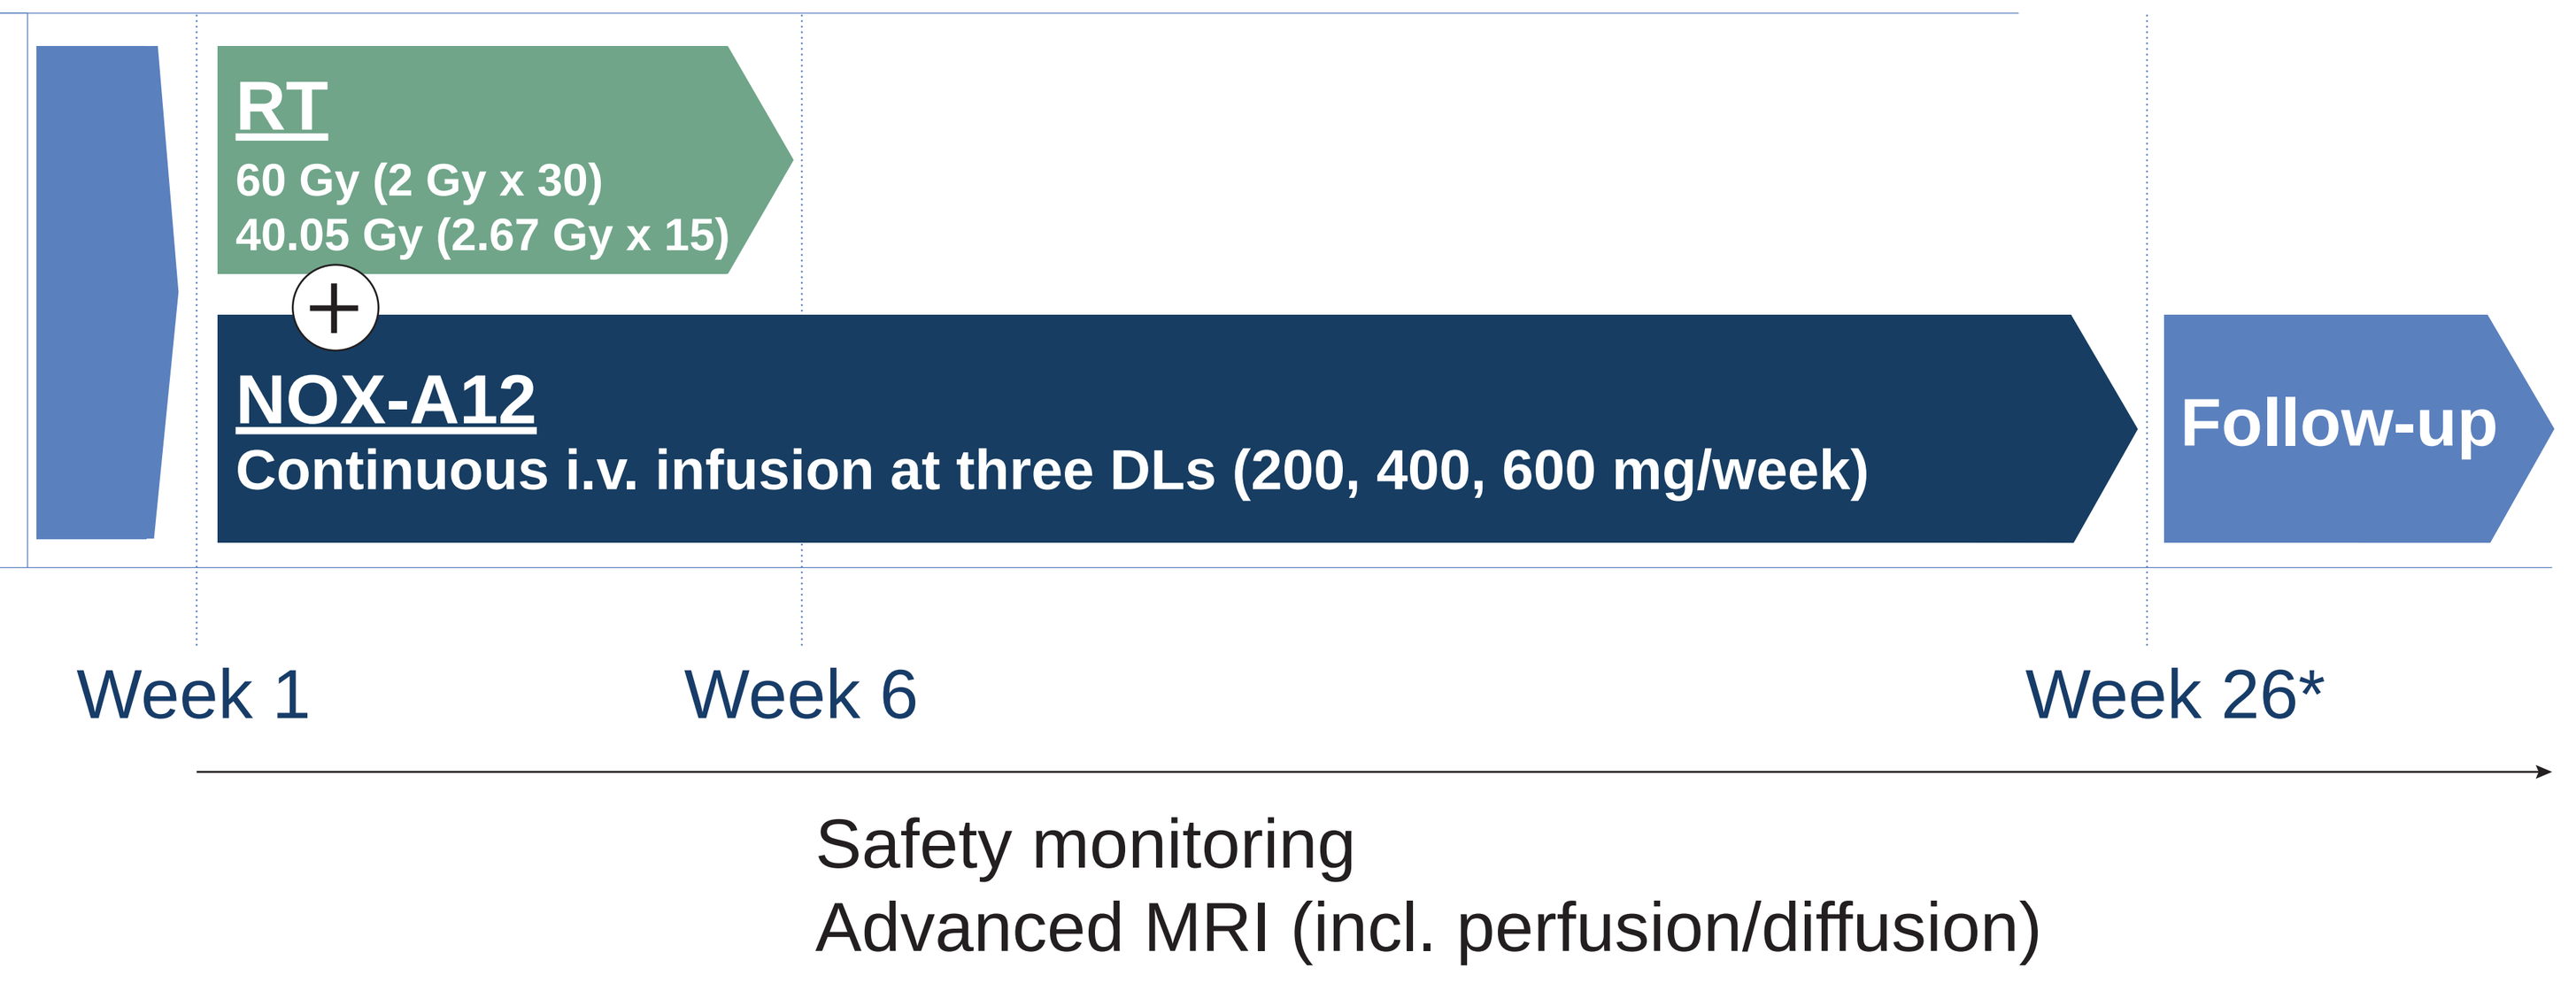
RT
60 Gy (2 Gy x 30)
40.05 Gy (2.67 Gy x 15)
+
NOX-A12
Continuous i.v. infusion at three DLs (200, 400, 600 mg/week)
Follow-up
Week 1
Week 6
Week 26*
Safety monitoring
Advanced MRI (incl. perfusion/diffusion)

Primary endpoint: Safety (incidence of adverse events)
Secondary endpoint: NOX-A12 plasma levels, radiographic response, PFS, OS
Exploratory endpoints: Multiplexed immunofluorescence imaging (CODEX®) - assessment of tissue biomarkers

- 10 patients with newly diagnosed incompletely resected (n=8) or biopsied (n=2) GBM with ECOG≤2, age≥18 and without MGMT promoter methylation were enrolled
- All patients received standard RT (60 Gy in 30 fractions or 40.05 Gy in 15 fractions) and escalating dose levels of continuous (24/7) i.v. infusions of NOX-A12 for 26 weeks

* End of treatment: 26 weeks; continuation beyond 26 weeks per investigator's decision

2 Trial outcomes

Patient characteristics (n=10)

Variable	n (%)	Median (range)
Gender		
Male	7 (70)	
Female	3 (30)	
Age (years)		65 (43-79)
ECOG Score		
0	7 (70)	
1	3 (30)	
Baseline NANO score		0 (0-4)
Resection status		
Incomplete	8 (80)	
Biopsy	2 (20)	
Residual volume (cc)		4.3 (2.5-34.1)
Weeks post surgery		4.6 (3.9-7.1)
Tumor localization		
Frontal lobe	4 (40)	
Temporal lobe	5 (50)	
Parietal lobe	3 (30)	
Occipital lobe	2 (20)	
Radiotherapy		
Normofractionated	7 (70)	
Hypofractionated	3 (30)	

Adverse Events

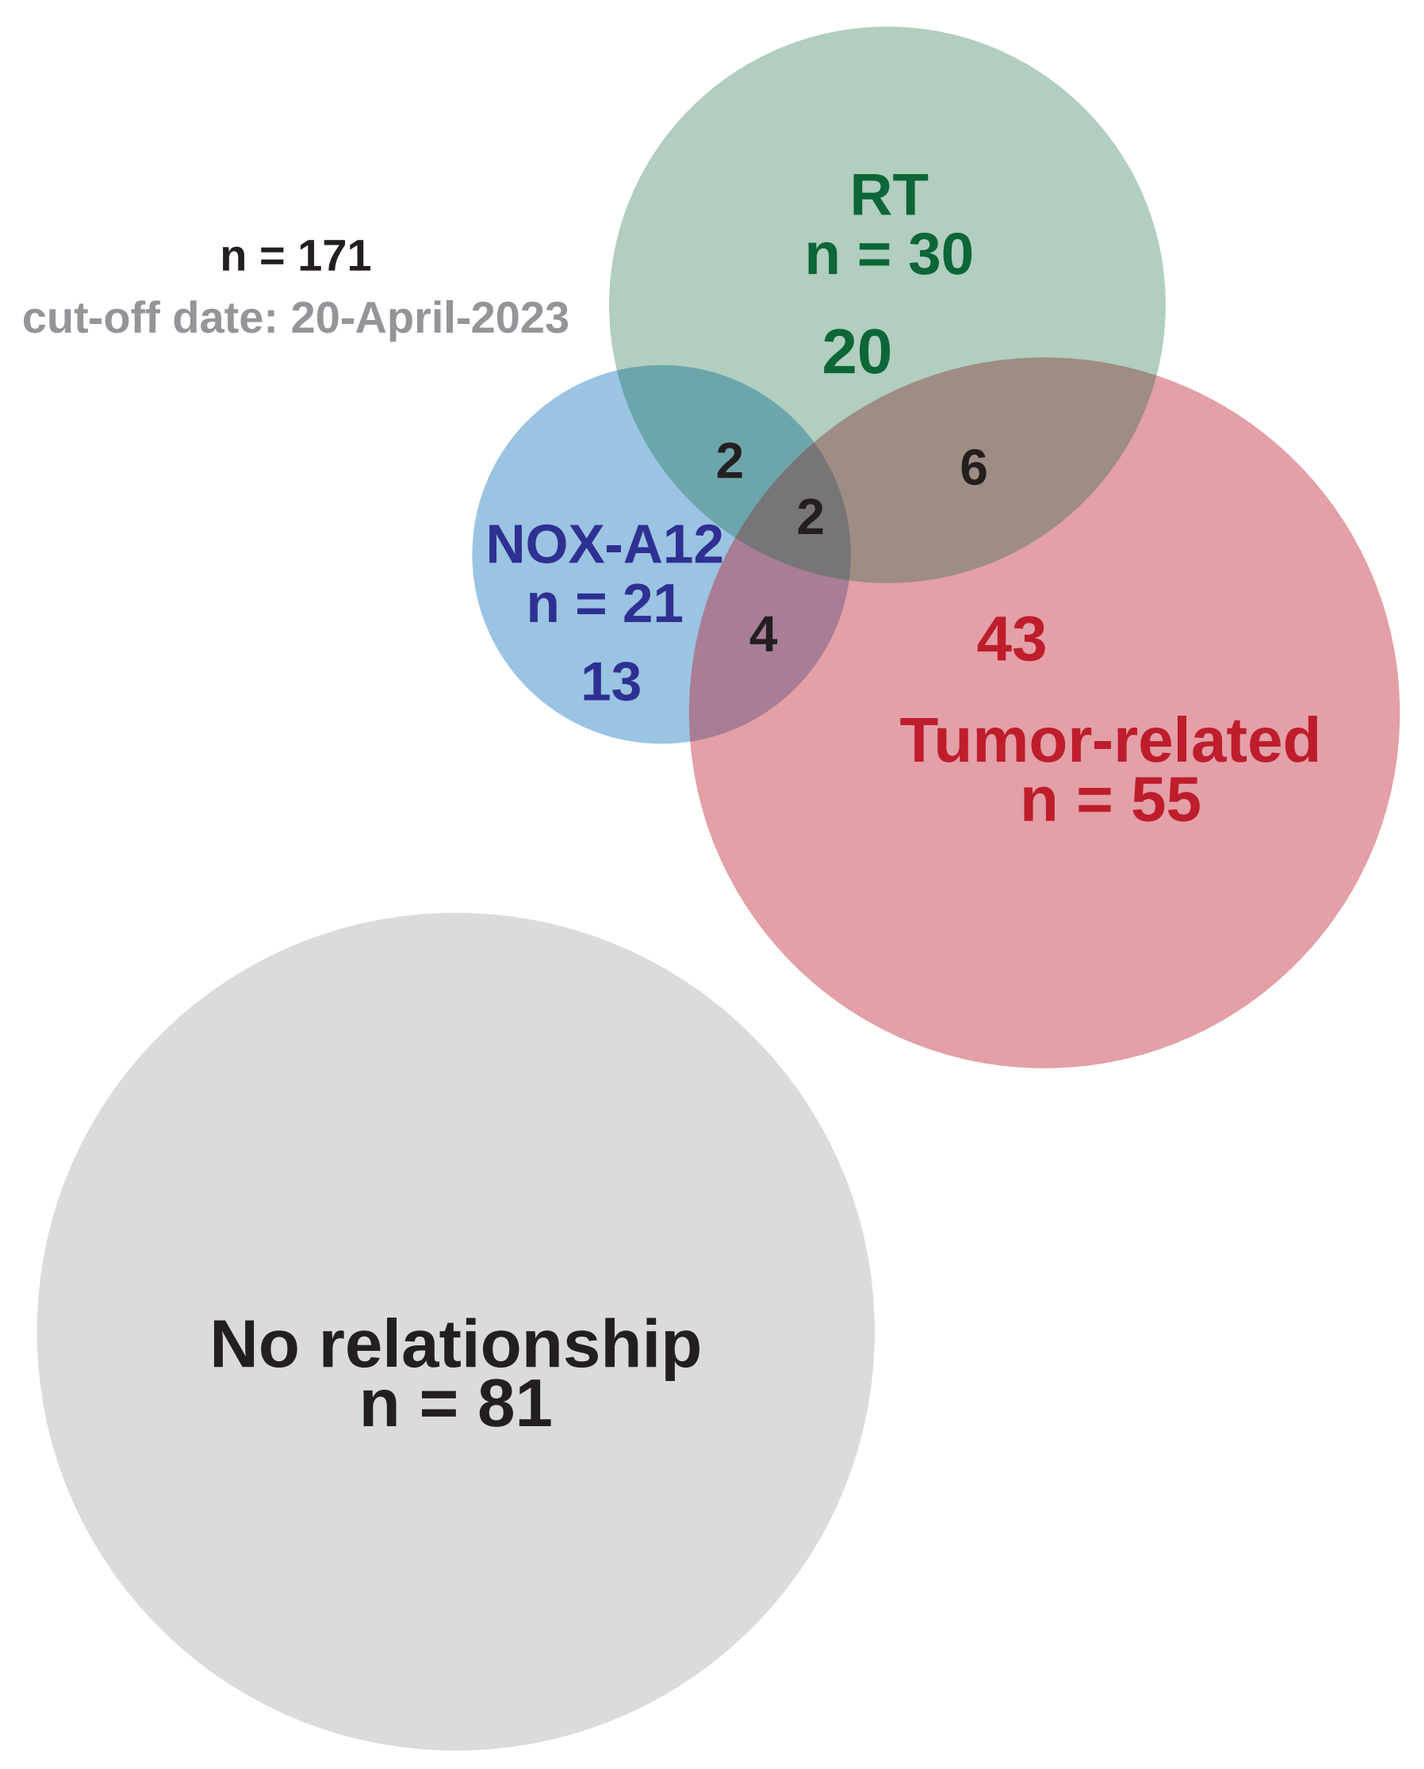
n = 171
cut-off date: 20-April-2023
RT
n = 30
20
NOX-A12
n = 21
13
2
2
6
4
43
Tumor-related
n = 55
No relationship
n = 81

Progression-free survival
GLORIA

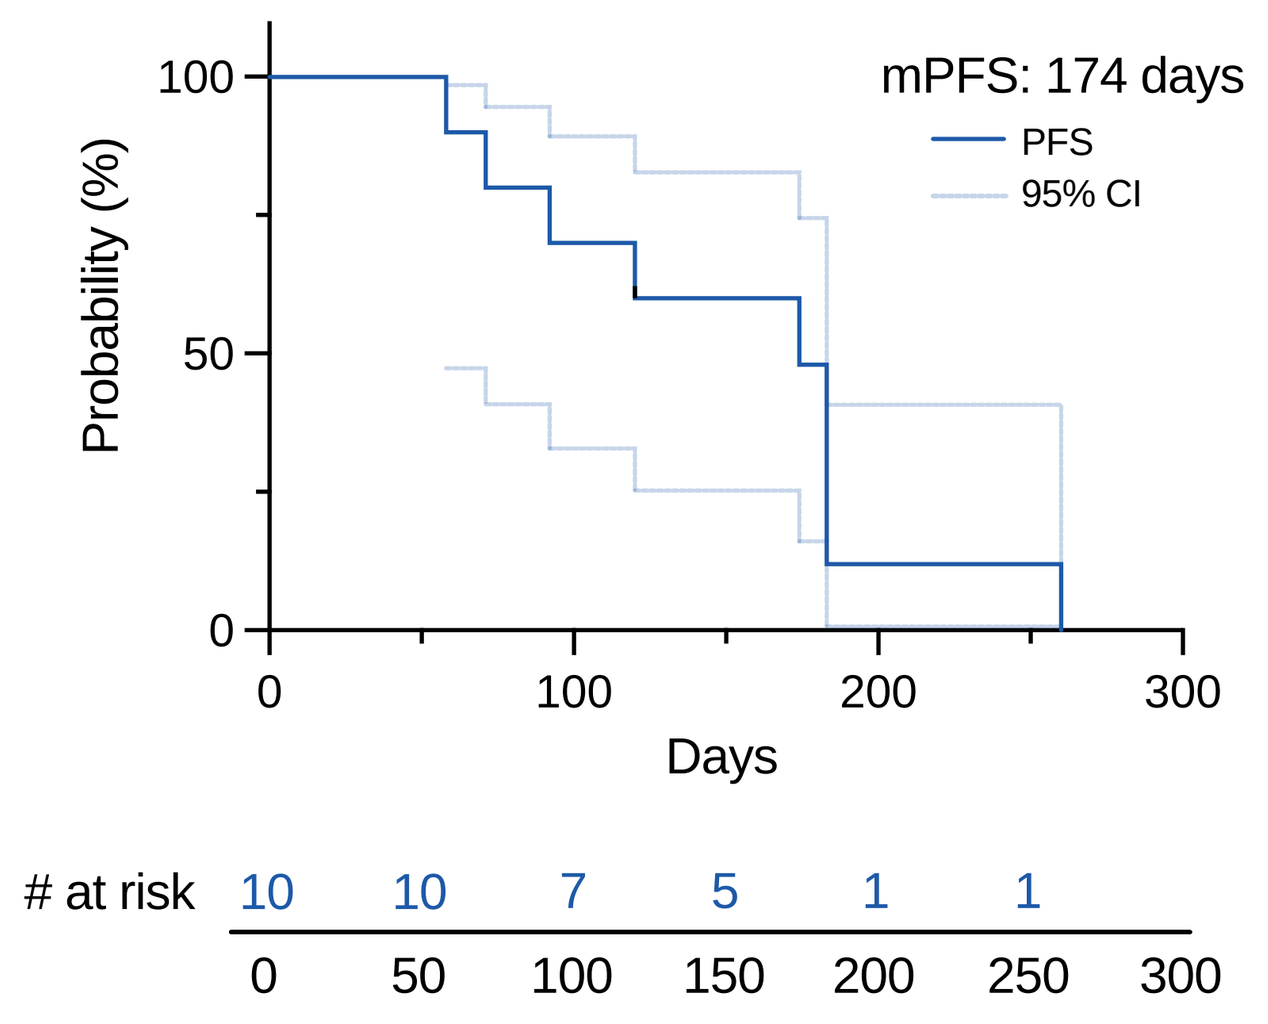
mPFS: 174 days
PFS
95% CI
Probability (%)
Days
at risk 10 10 7 5 1 1
0 50 100 150 200 250 300

Overall survival
GLORIA

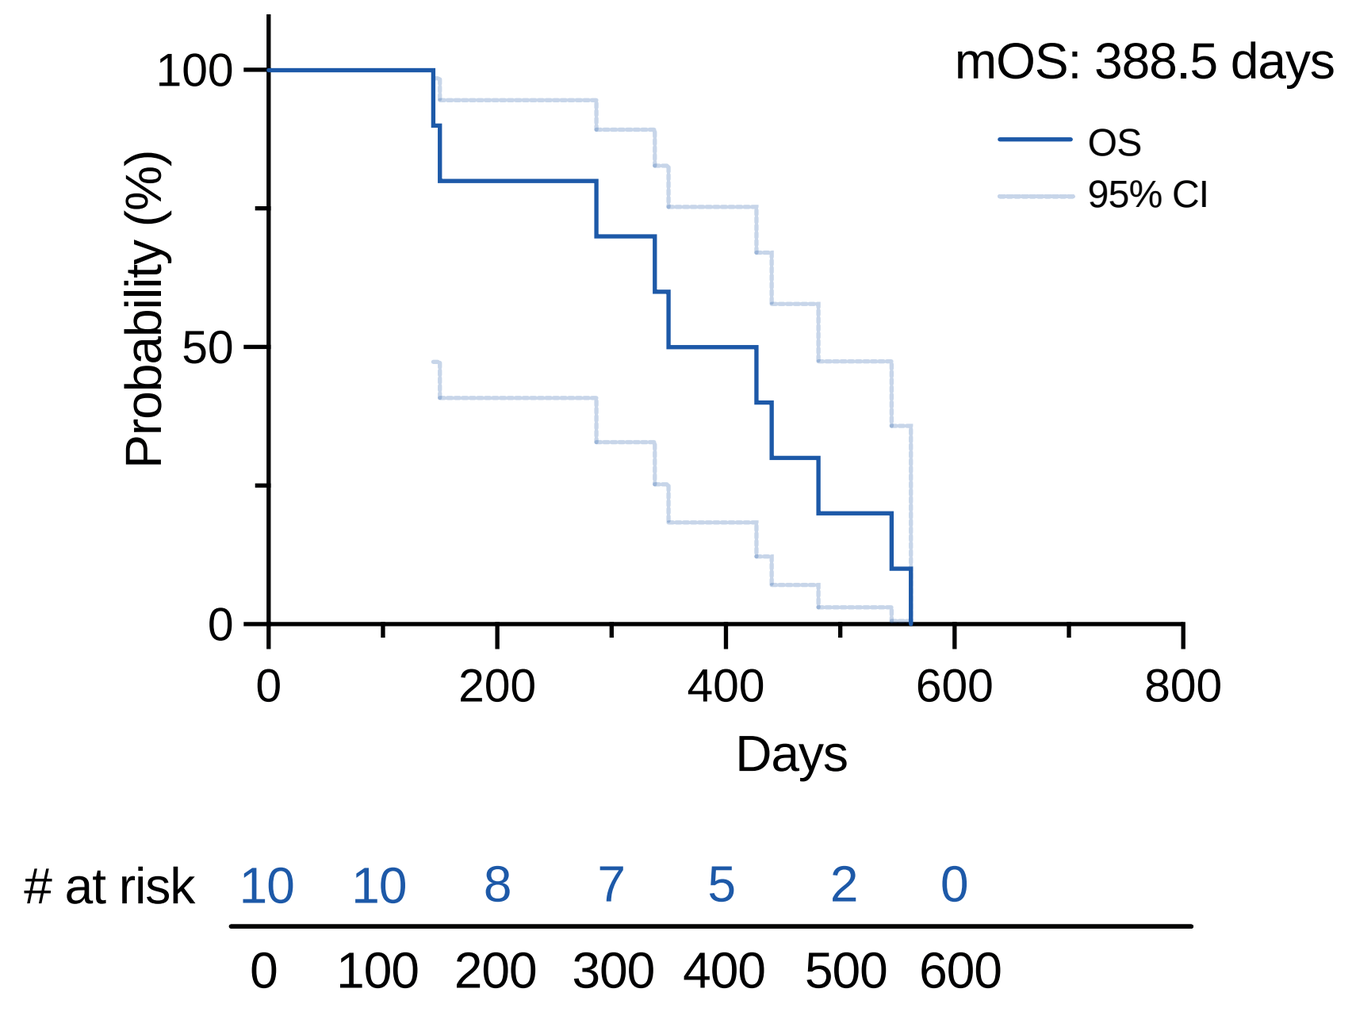
mOS: 388.5 days
OS
95% CI
Probability (%)
Days
at risk 10 10 8 7 5 2 0
0 100 200 300 400 500 600

- Treatment was safe with no dose-limiting toxicities
- Of all G≥2 adverse events (n=84), 4 (4.7%) were considered to be solely NOX-A12-related, including 1 G3 AE at DL 3 (elevation of GGT)
- Median PFS of the GLORIA cohort was 5.7 (range 1.9–8.5) months and the median OS was 12.7 (4.7–18.4) months

3 scRNAseq and tissue analysis

CXCL12 expression in scRNAseq from human GBM
Abdelfattah et al. 2022

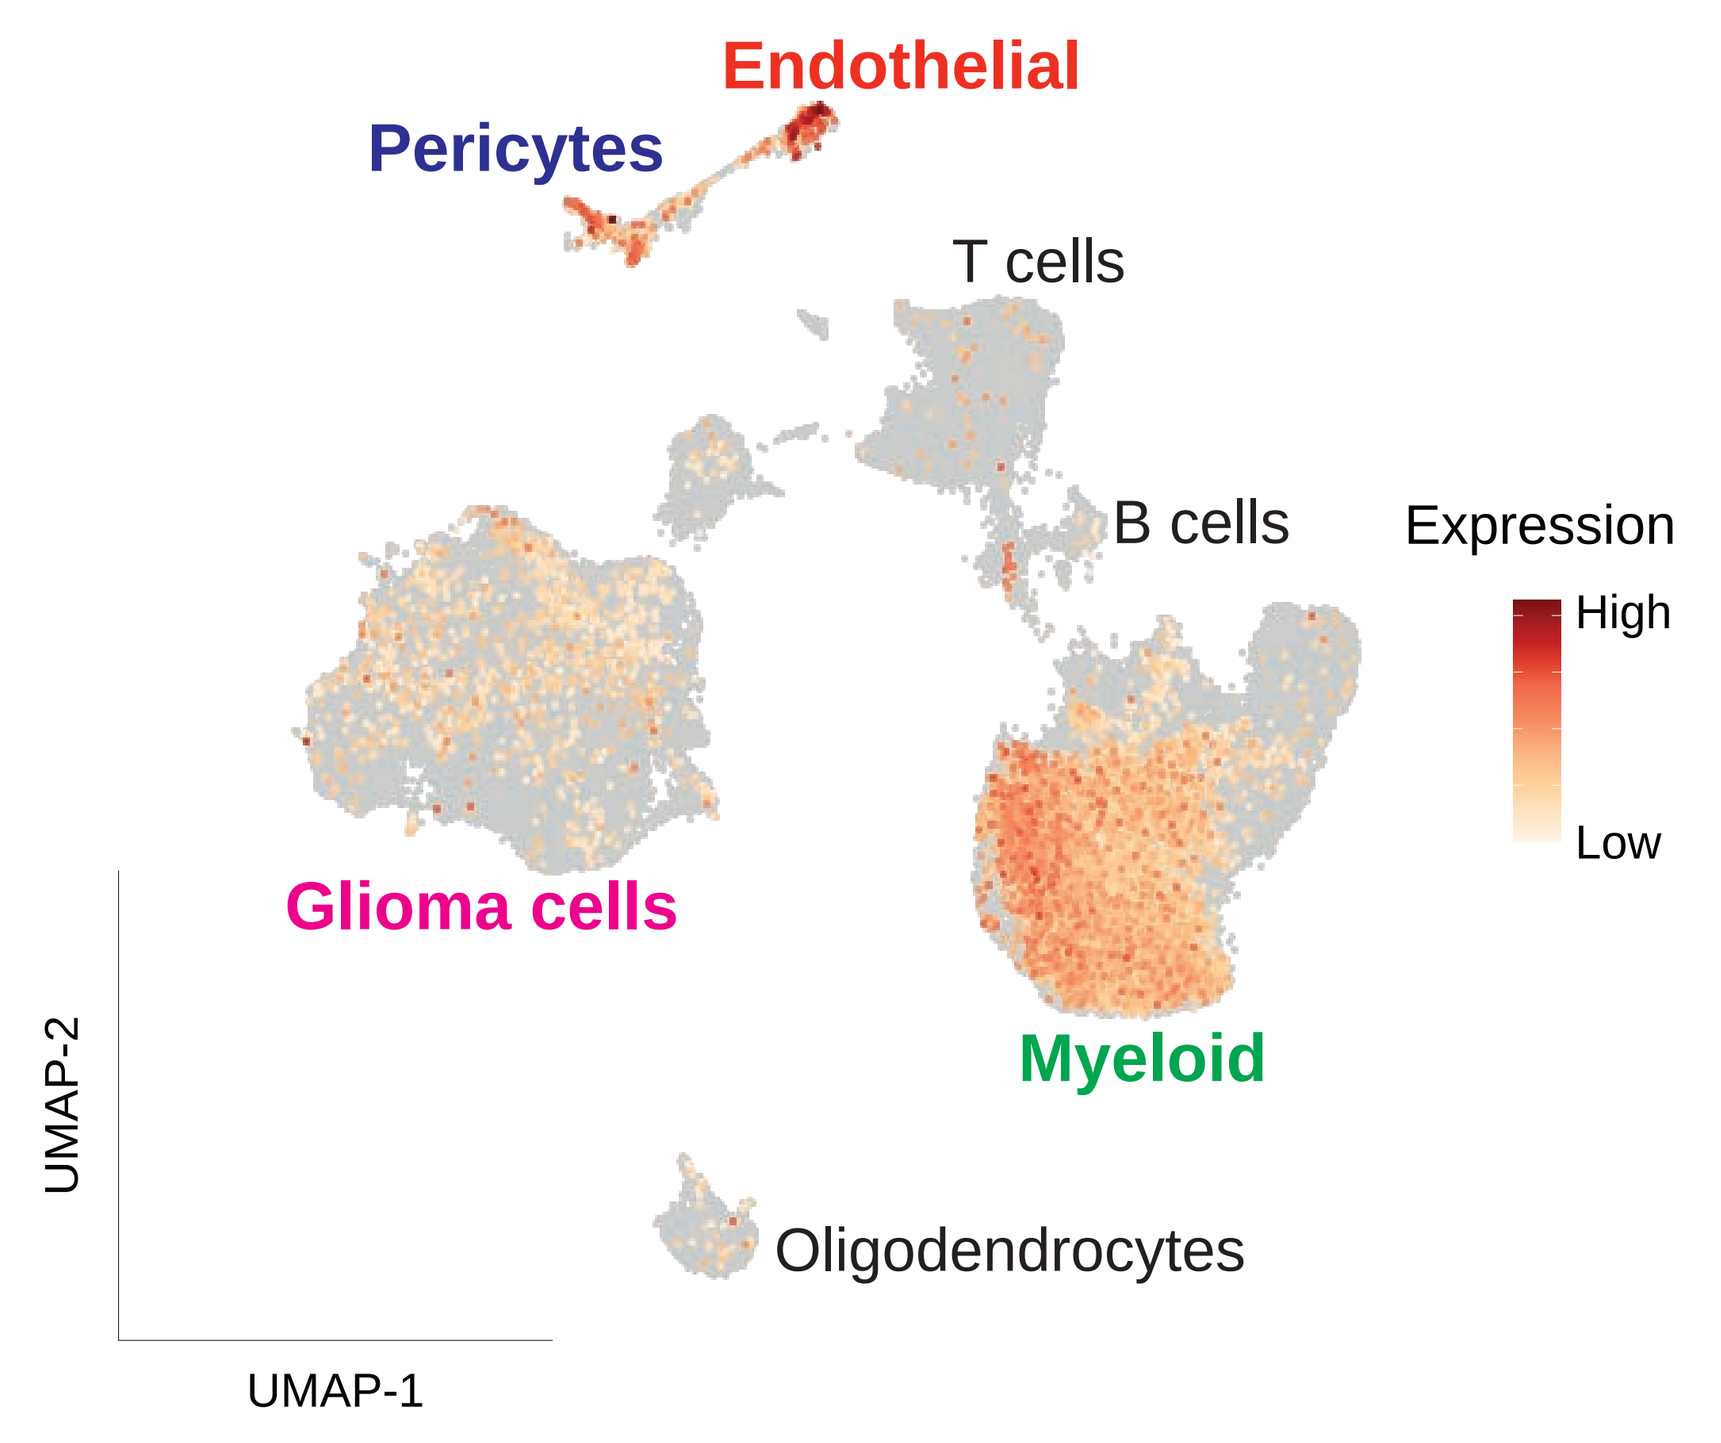
Endothelial
Pericytes
T cells
B cells
Expression
High
Low
Glioma cells
Myeloid
Oligodendrocytes
UMAP-2
UMAP-1

- Highest CXCL12 mRNA expression in endothelial cells, followed by pericytes, myeloid (macrophages, microglia) and glioma cells

Workflow

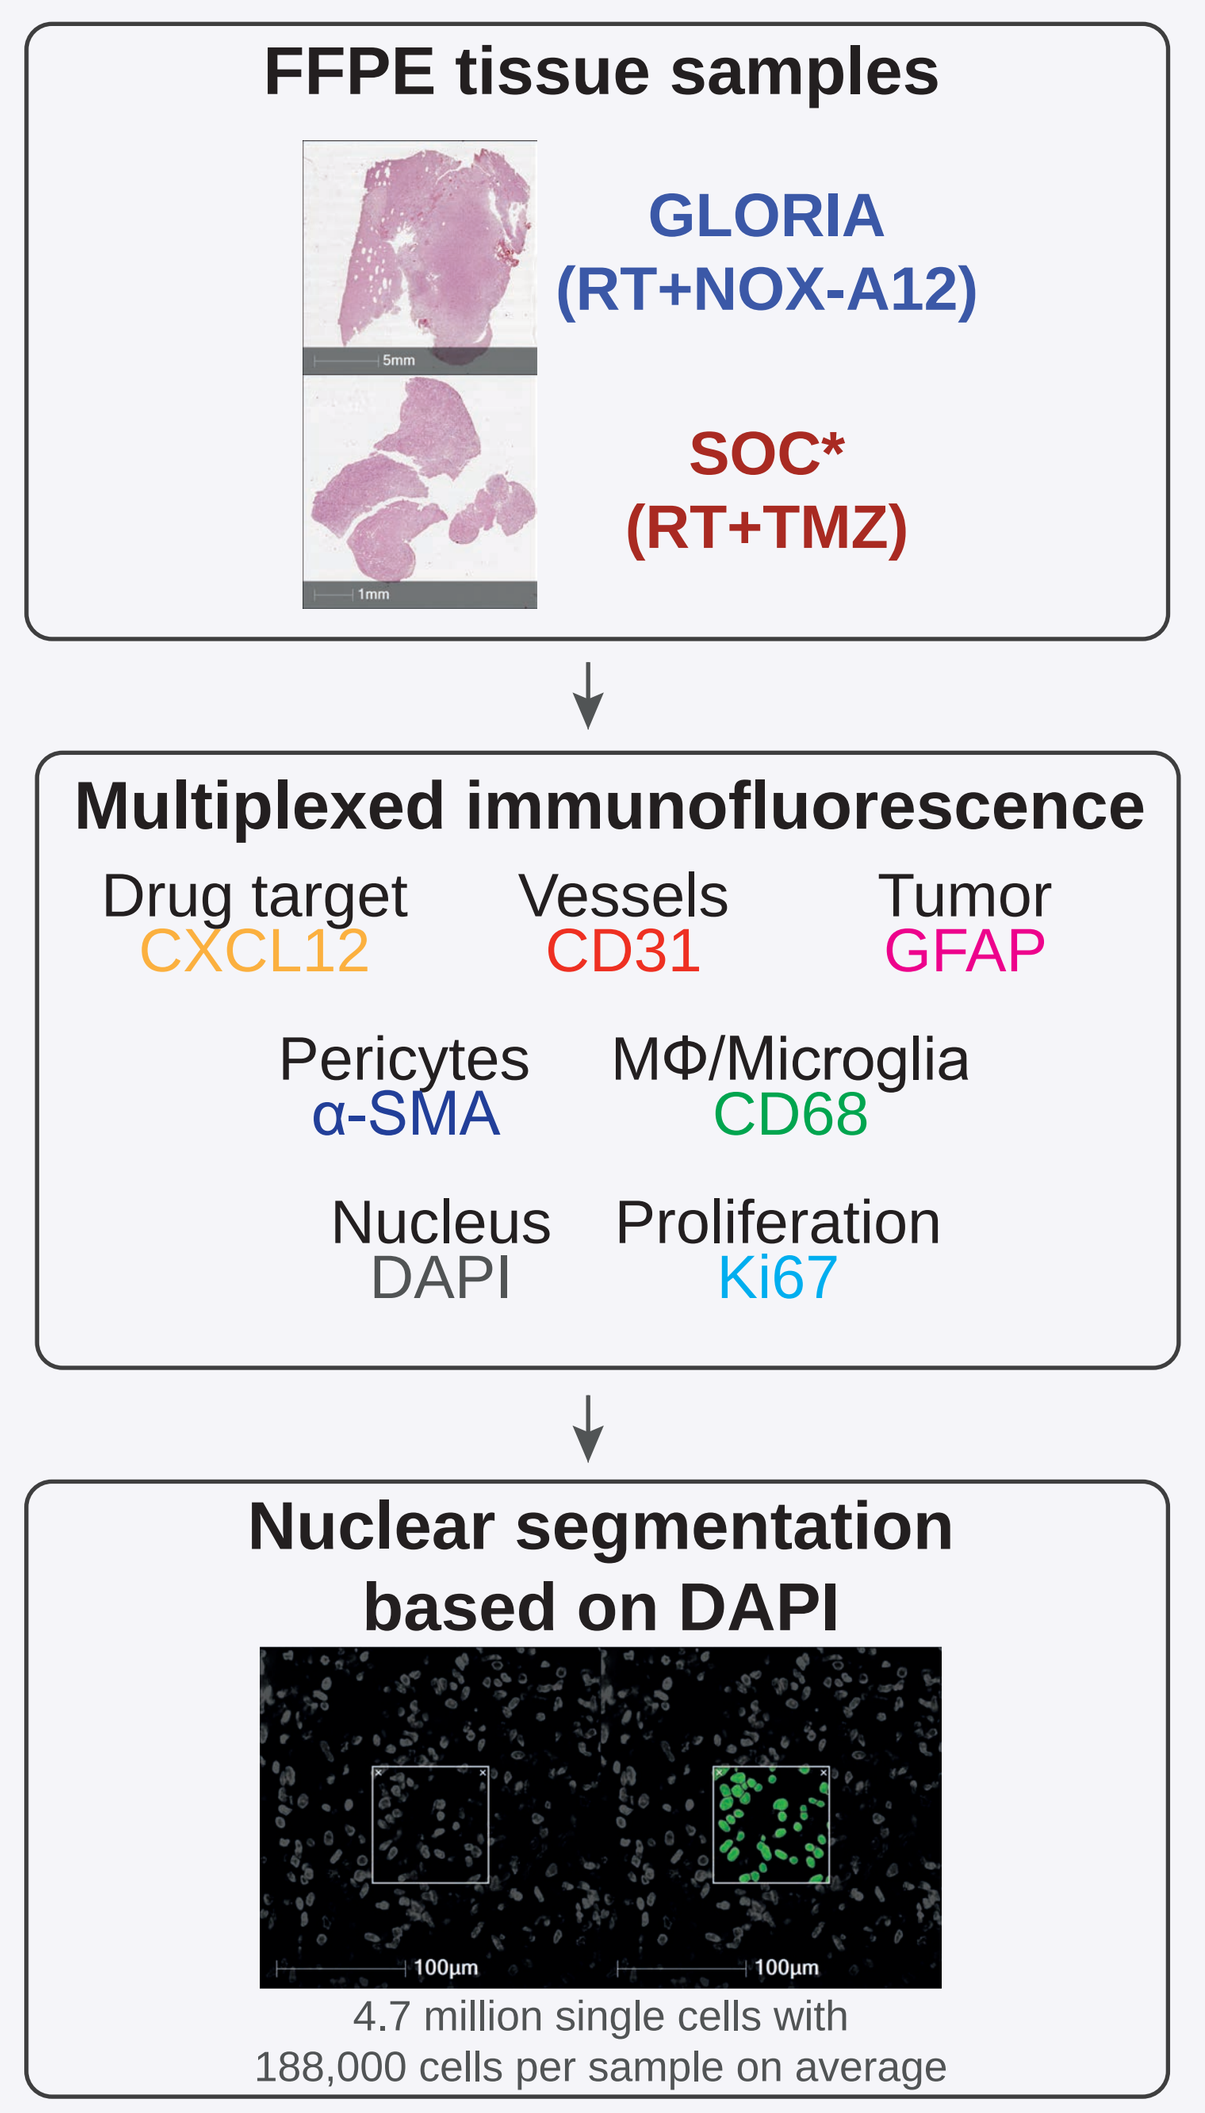
FFPE tissue samples
GLORIA
(RT+NOX-A12)
SOC*
(RT+TMZ)
Multiplexed immunofluorescence
Drug target
CXCL12
Vessels
CD31
Tumor
GFAP
Pericytes
α-SMA
MΦ/Microglia
CD68
Nucleus
DAPI
Proliferation
Ki67
Nuclear segmentation based on DAPI
4.7 million single cells with
188,000 cells per sample on average

Multiplexed immunofluorescence for CXCL12 positivity

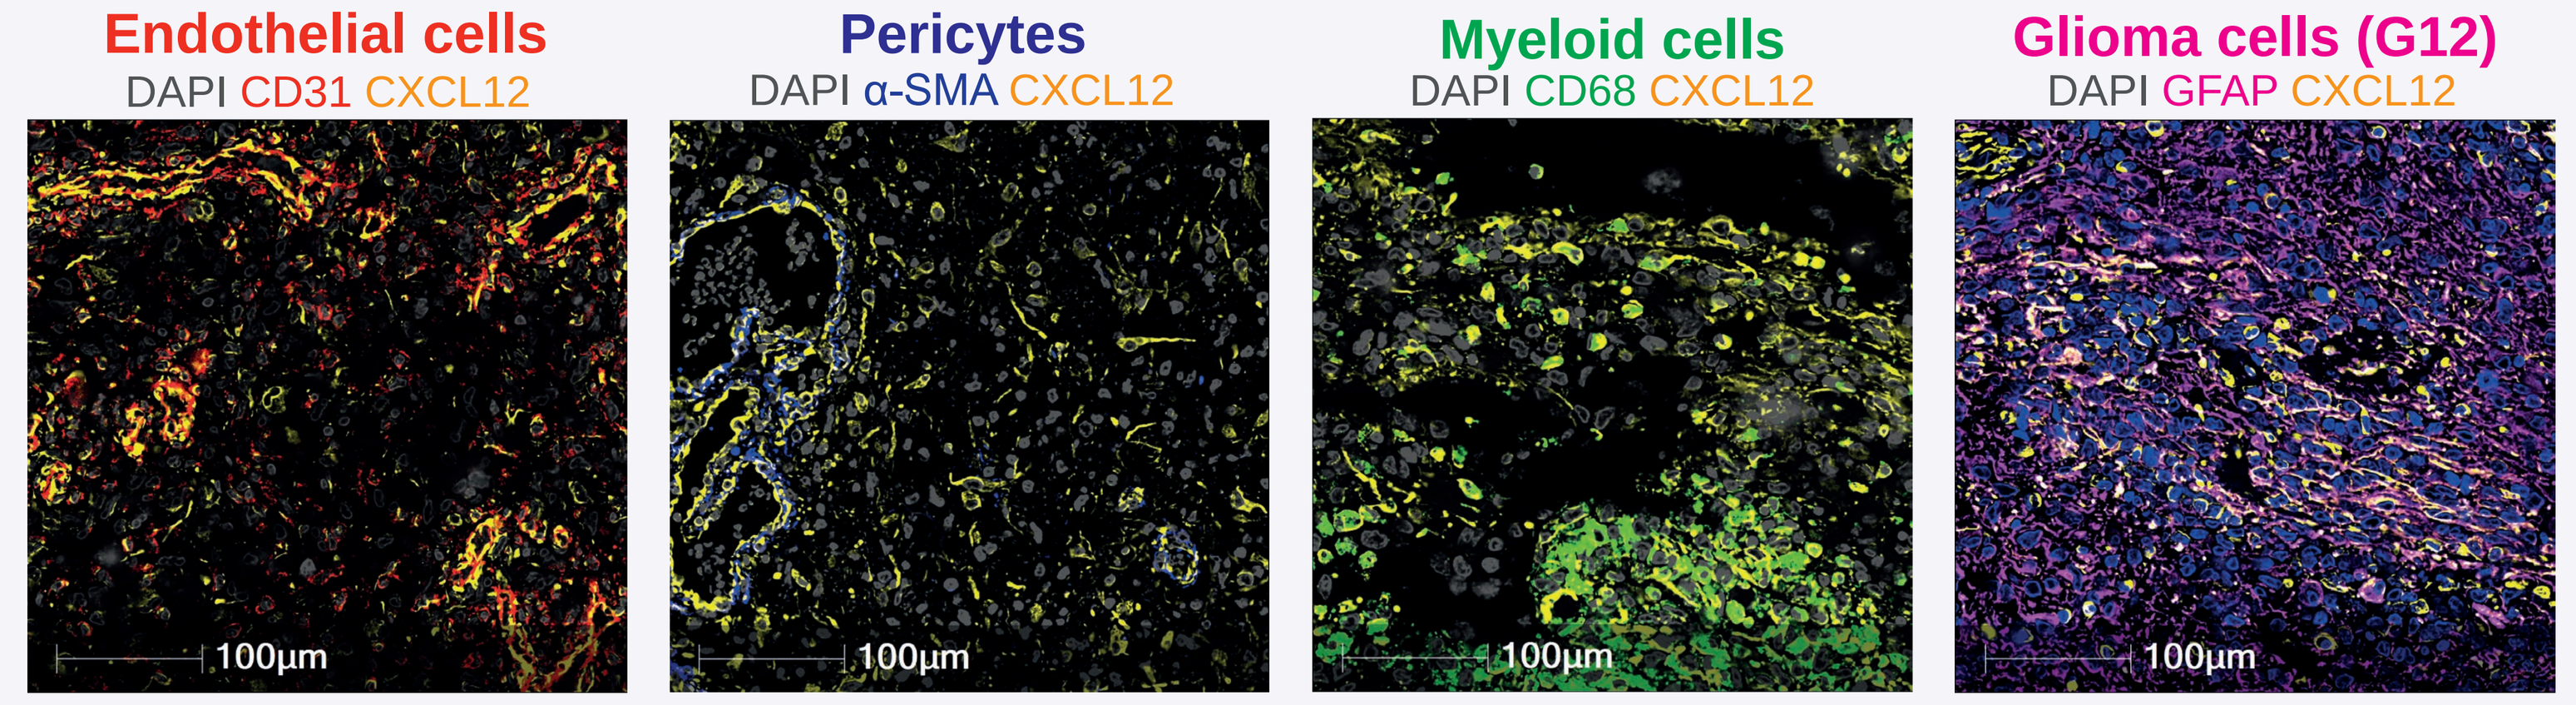
Endothelial cells
DAPI CD31 CXCL12
Pericytes
DAPI α-SMA CXCL12
Myeloid cells
DAPI CD68 CXCL12
Glioma cells (G12)
DAPI GFAP CXCL12
100µm
100µm
100µm
100µm

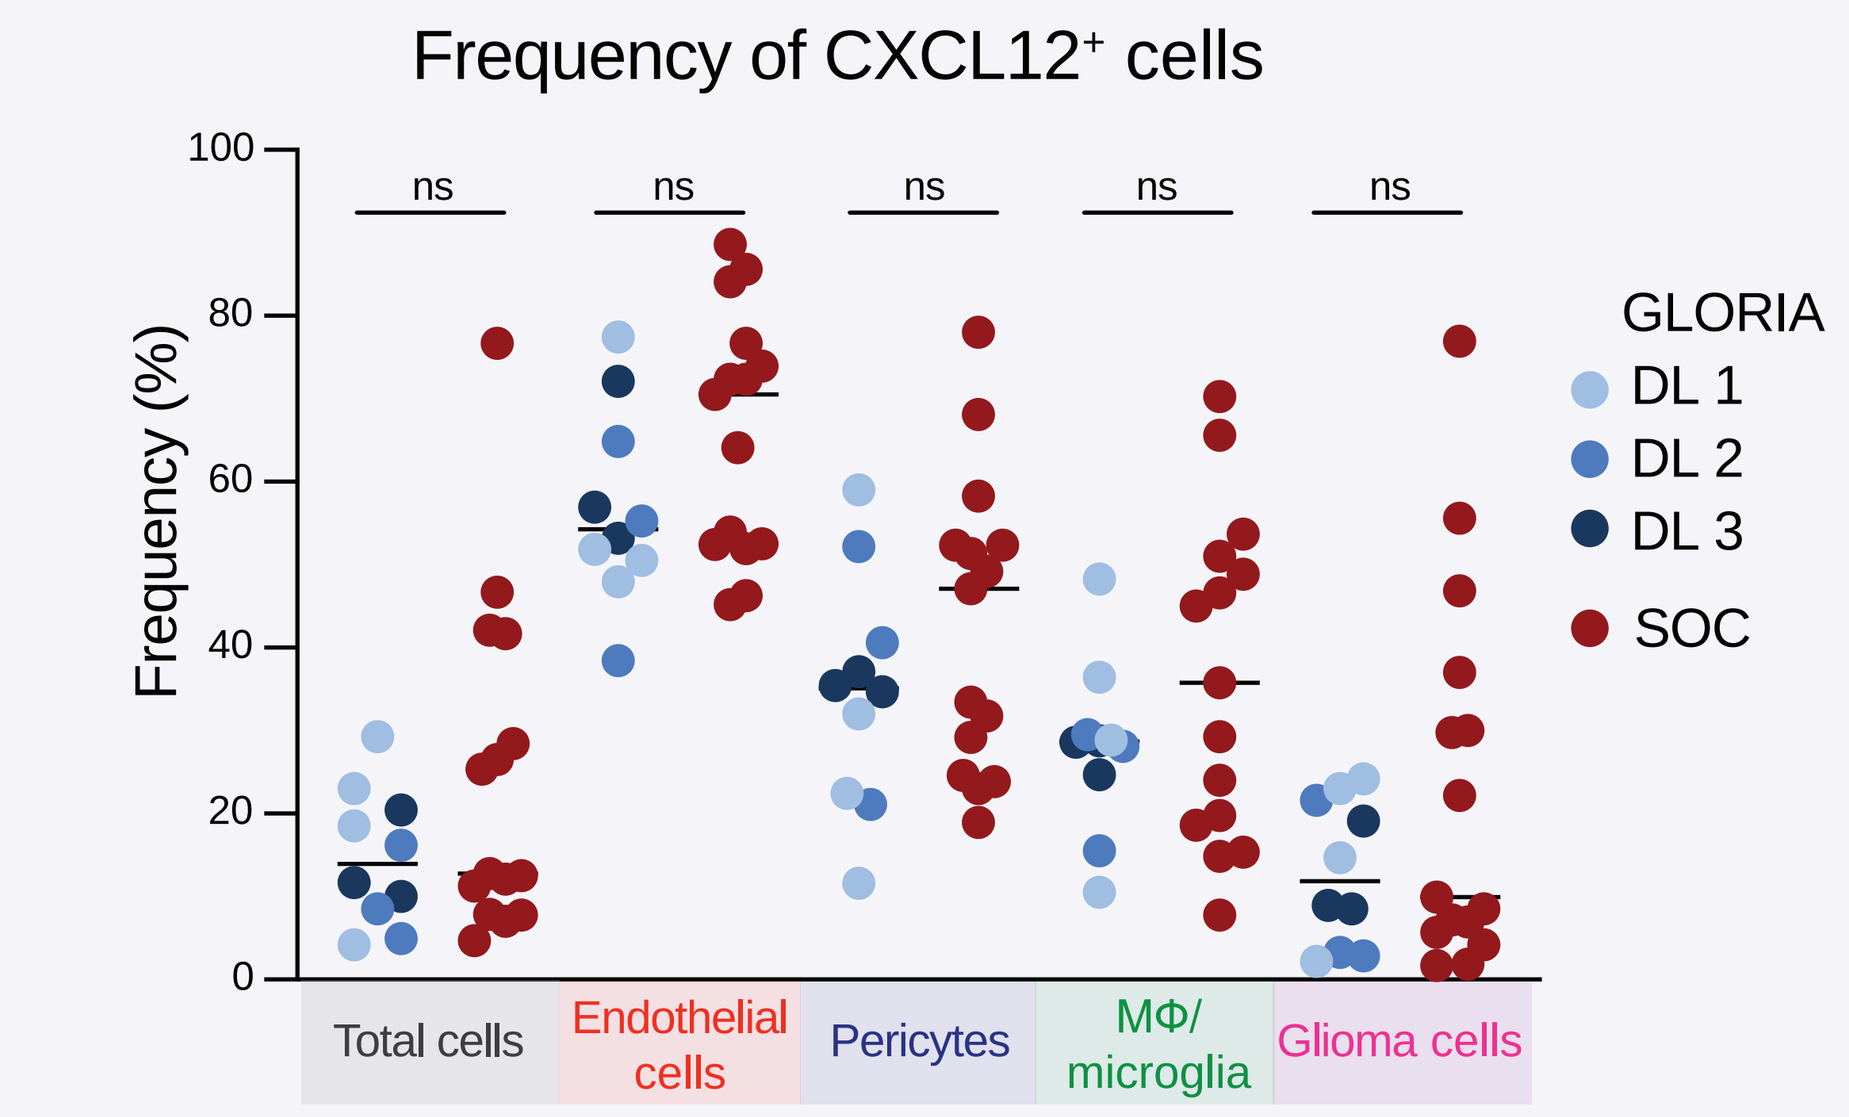
Frequency of CXCL12+ cells
Frequency (%)
ns
ns
ns
ns
ns
GLORIA
DL 1
DL 2
DL 3
SOC
Total cells
Endothelial cells
Pericytes
MΦ/ microglia
Glioma cells

- Cell-type specific CXCL12 protein expression assessed in pre-treatment tumor samples from:
 - GLORIA patients (n=10)
 - SOC cohort (n=15)*
- Tissue analysis confirms cell type-specific expression of CXCL12 seen in scRNAseq
- No signifcant differences in CXCL12+ cell frequency between GLORIA and SOC

*SOC cohort: external control with incompletely resected or biopsied GBM (WHO CNS grade 4) lacking MGMT promoter methylation and treated with standard of care (SOC) (n=15).

High frequency of CXCL12+ endothelial / glioma cells is predictive for clinical efficacy of RT + CXCL12-inhibition in GBM

4 CXCL12 positivity and PFS

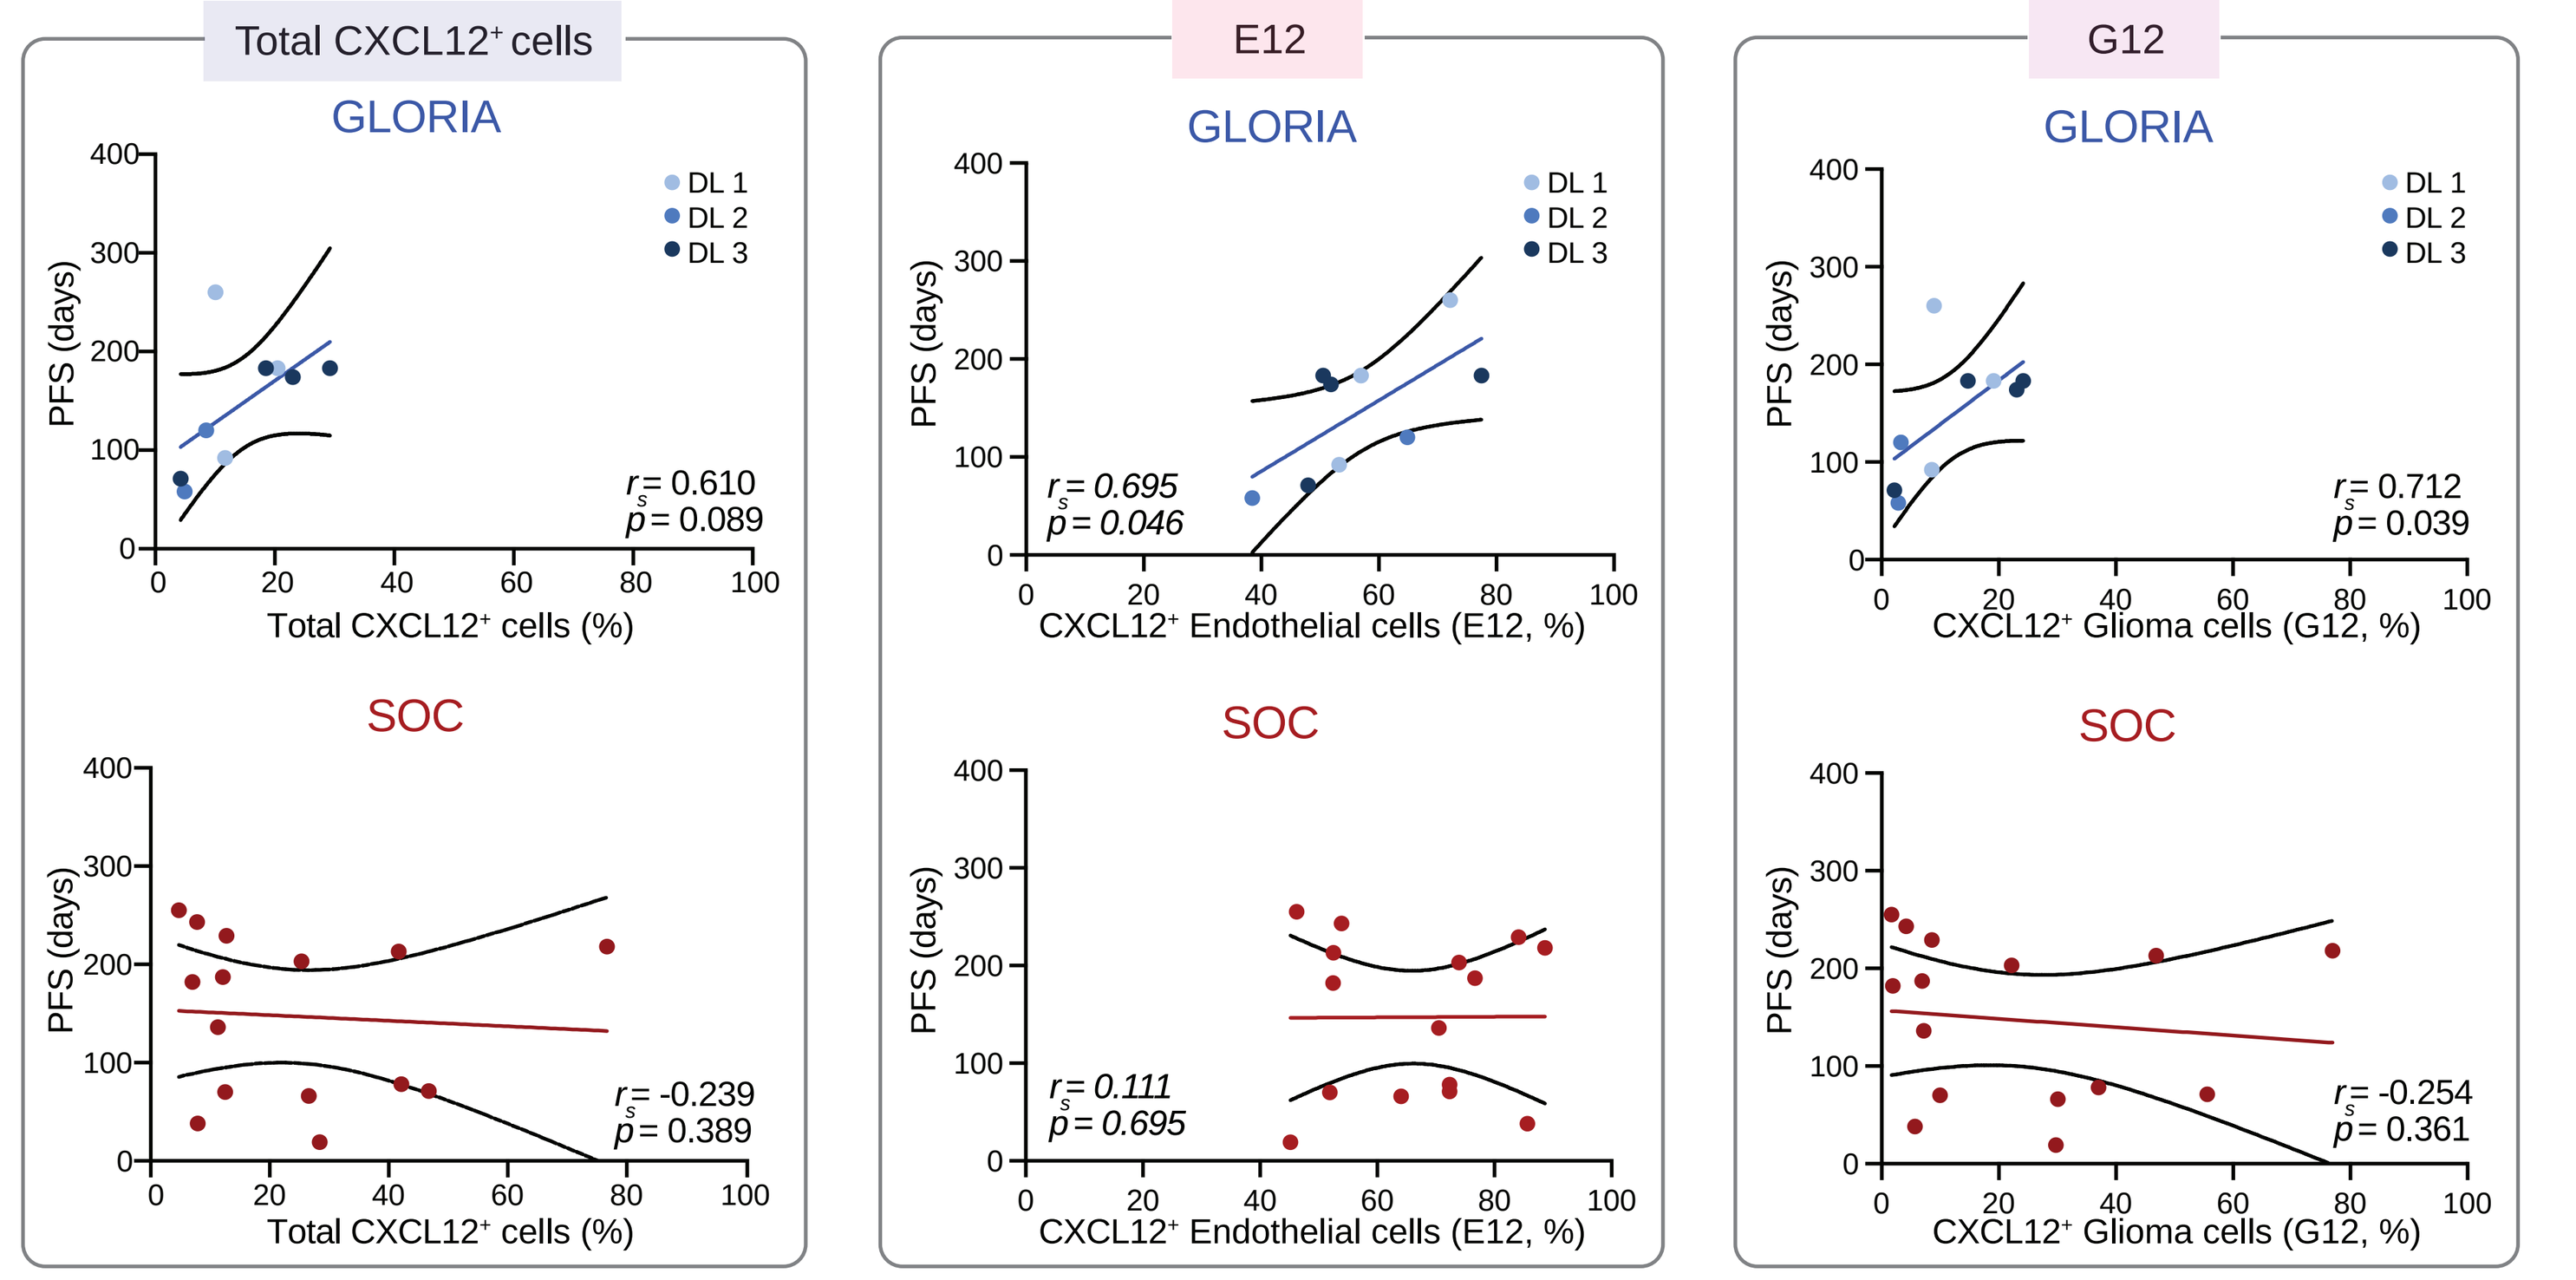
Total CXCL12+ cells
GLORIA
r_s = 0.610
p = 0.089
Total CXCL12+ cells (%)
SOC
r_s = -0.239
p = 0.389
E12
GLORIA
r_s = 0.695
p = 0.046
CXCL12+ Endothelial cells (E12, %)
SOC
r_s = 0.111
p = 0.695
G12
GLORIA
r_s = 0.712
p = 0.039
CXCL12+ Glioma cells (G12, %)
SOC
r_s = -0.254
p = 0.361
PFS (days)
DL 1
DL 2
DL 3

- Positive trend between frequency of total CXCL12+ cells and PFS in the GLORIA cohort
- Positive correlation between CXCL12+ endothelial cells (E12) and CXCL12+ glioma cells (G12) with PFS in the GLORIA cohort
- No correlations or trend in the SOC cohort

5 EG12 score

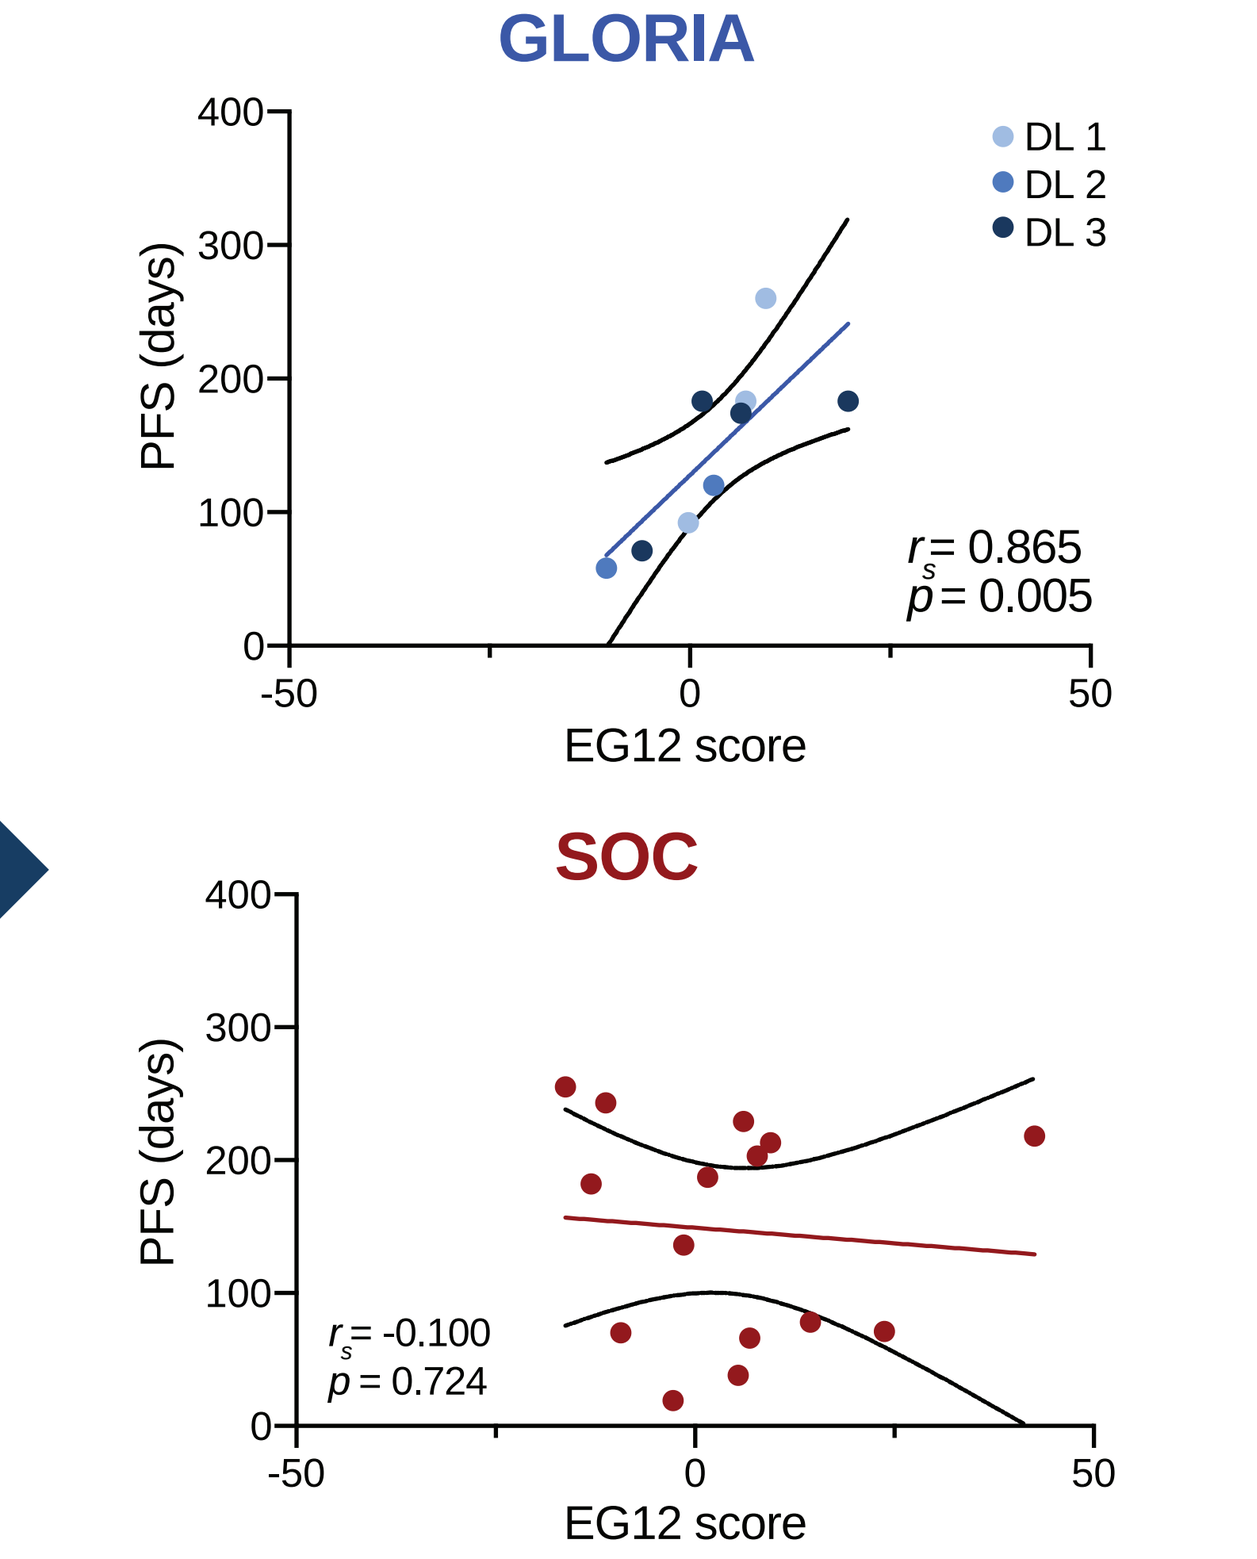
GLORIA
DL 1
DL 2
DL 3
PFS (days)
r_s = 0.865
p = 0.005
EG12 score
SOC
r_s = -0.100
p = 0.724
EG12 score

- Creation of a combined EG12 score incorporating endothelial and glioma cells (mean of the median-centered values of E12 and G12)

6 Stratified Survival

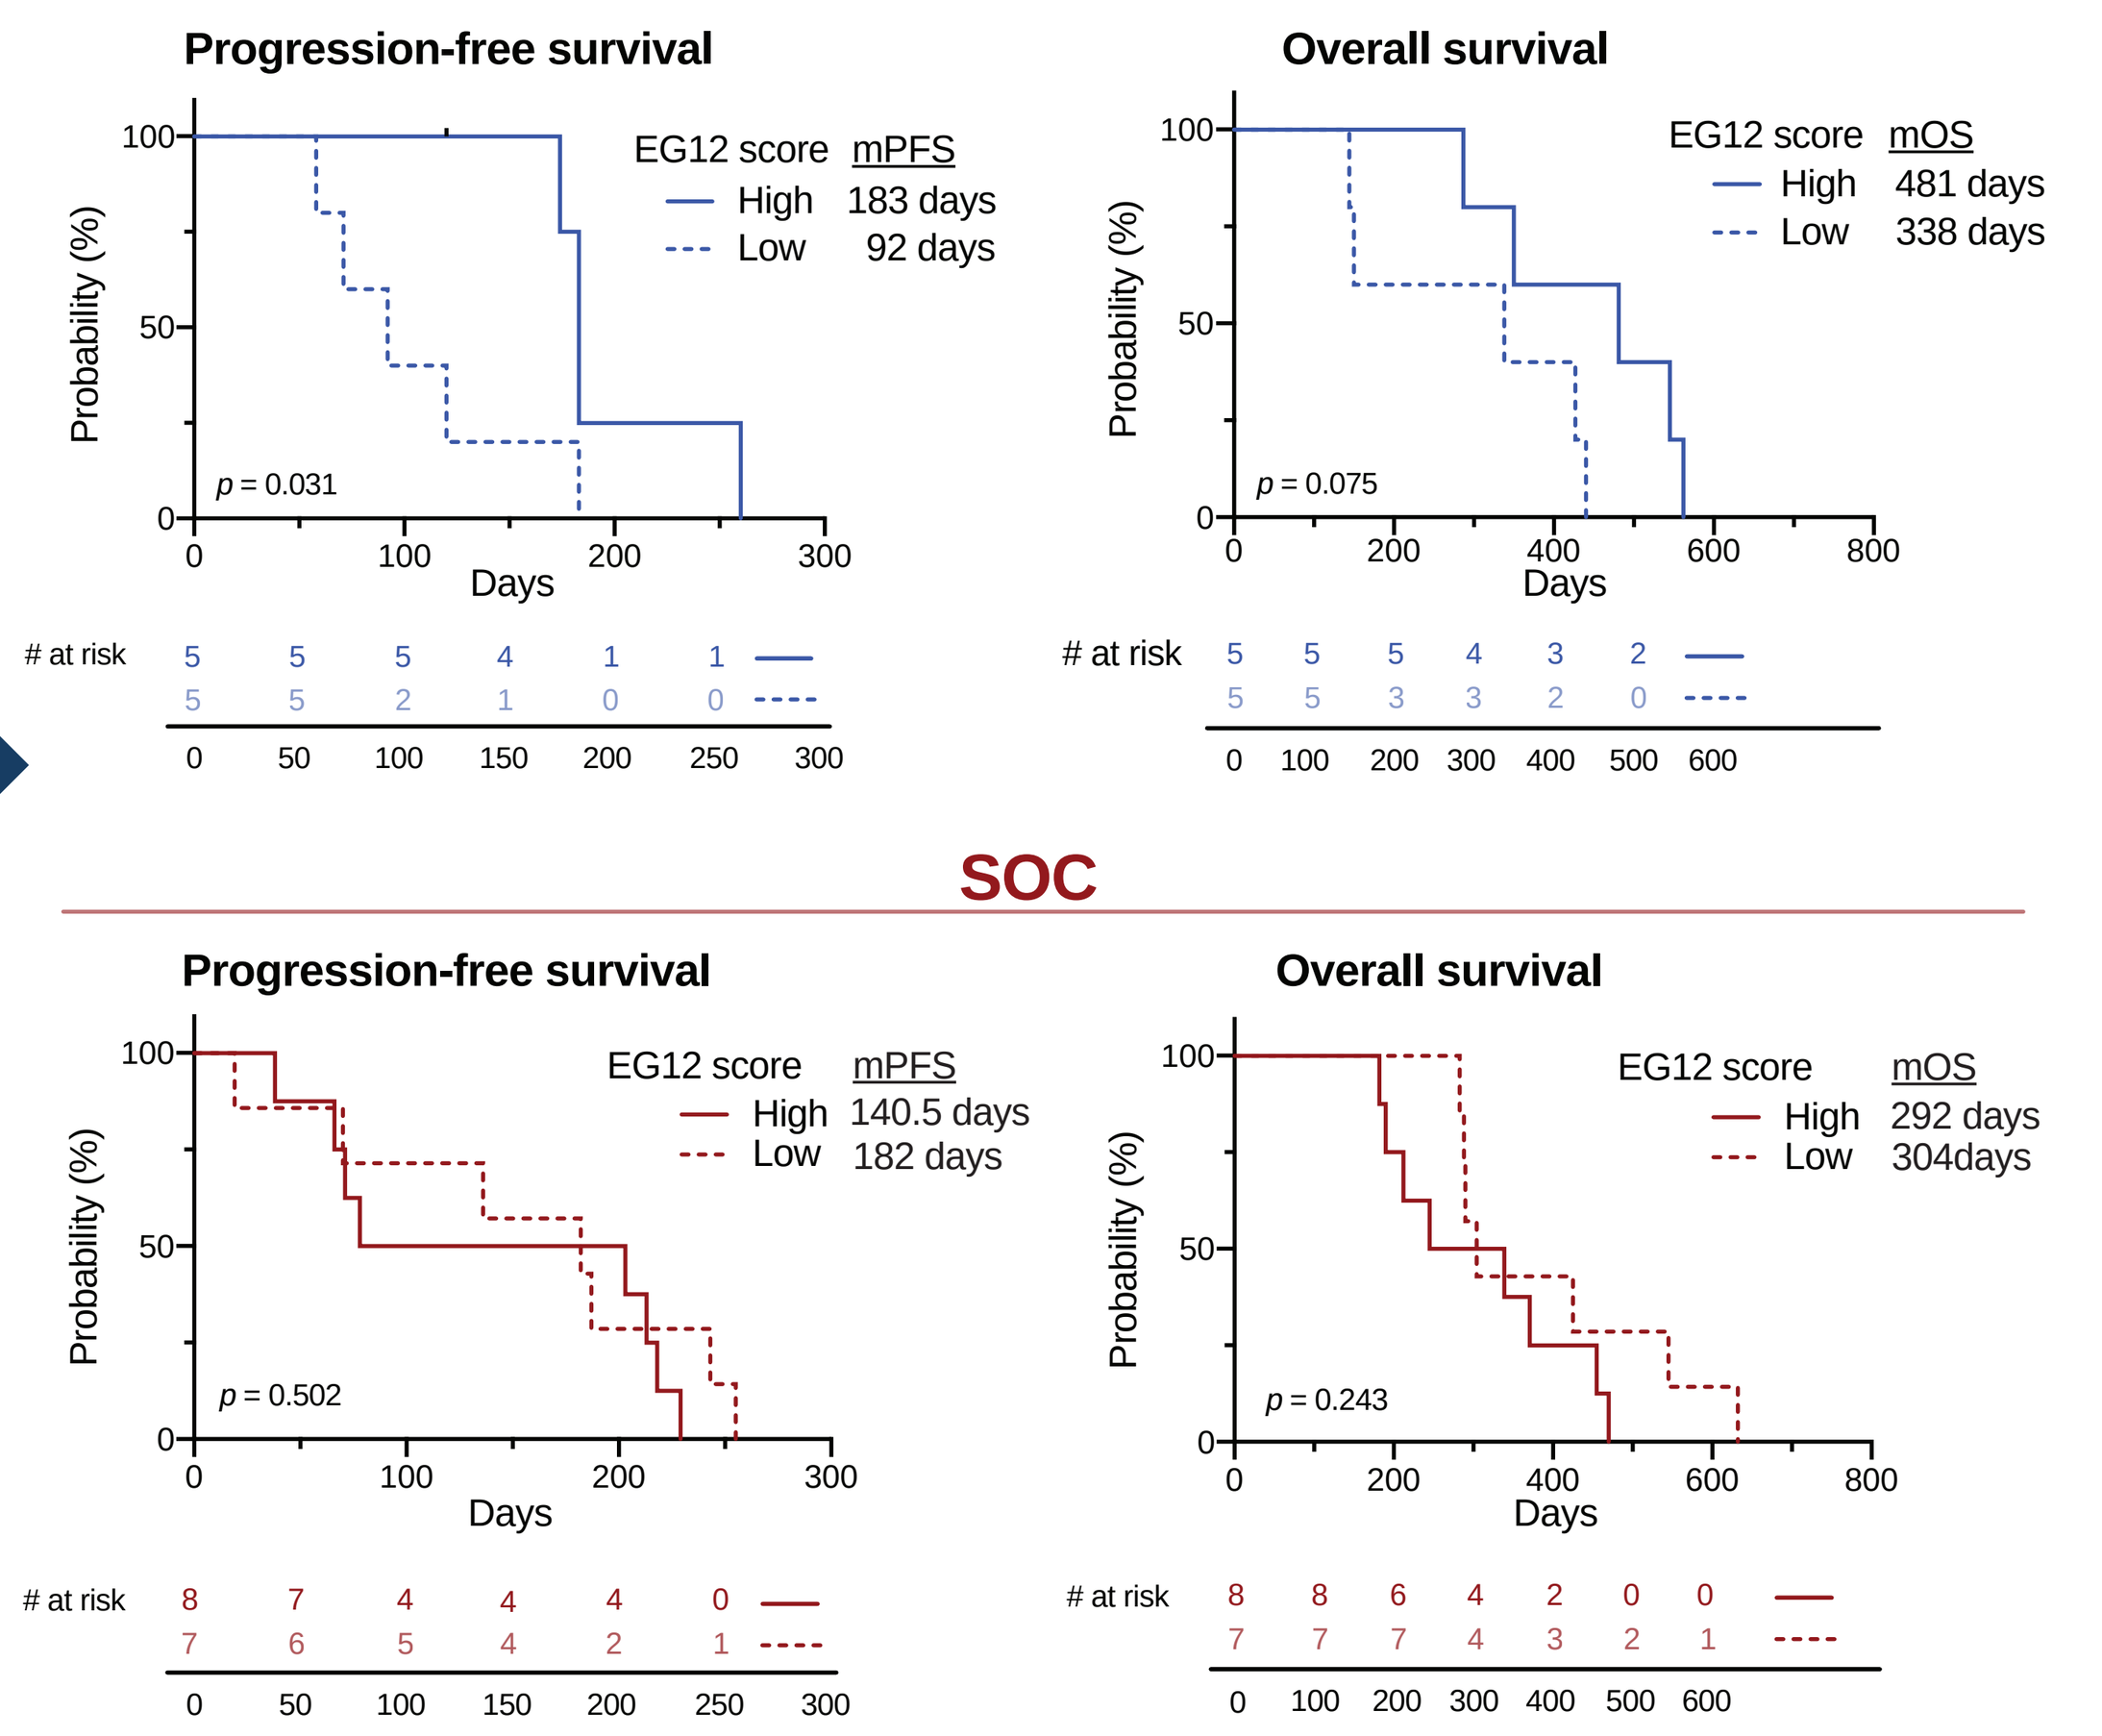
GLORIA
Progression-free survival
EG12 score mPFS
High 183 days
Low 92 days
p = 0.031
Overall survival
EG12 score mOS
High 481 days
Low 338 days
p = 0.075
SOC
Progression-free survival
EG12 score mPFS
High 140.5 days
Low 182 days
p = 0.502
Overall survival
EG12 score mOS
High 292 days
Low 304days
p = 0.243
Probability (%)
Days
at risk

* authors contributed equally

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the authors of this poster.