CXCL12 Inhibition with NOX-A12 (Olaptesed Pegol) Increases T and NK Cell Infiltration and Synergizes with Immune Checkpoint Blockade and ADCC in Tumour-Stroma Spheroids

Dirk Zboralski, Anna Kruschinski, Dirk Eulberg and Axel Vater

NOXXON | PHARMA AG

BACKGROUND & RATIONALE

T and NK cell-based cancer immunotherapy requires physical contact between immune effector cells and malignant cells, which is generally restricted by the tumour microenvironment (TME).[1] The chemokine CXCL12 has recently been described as a T cell exclusion factor in the TME-driven immune suppression.[2] The clinical stage L-aptamer (Spiegelmer®) NOX-A12 (olaptesed pegol) was found to detach CXCL12 from the surface of stromal cells[3] and to block CXCL12 binding to CXCR4 and CXCR7[4,5]. In this study we aimed to investigate whether NOX-A12 is able to enhance T and NK cell infiltration into tumour-stroma spheroids, thereby facilitating effective cancer immunotherapy.

Further known modes of action of the CXCL12 inhibitor NOX-A12:

- NOX-A12 mobilizes healthy immune cells [6, Figure 1]
- NOX-A12 exposes hidden tumour cells better to anti-cancer therapy by depriving their contact with the TME [3,4,7,8]
- NOX-A12 inhibits the formation and growth of metastases [4,9]

T cell mobilization

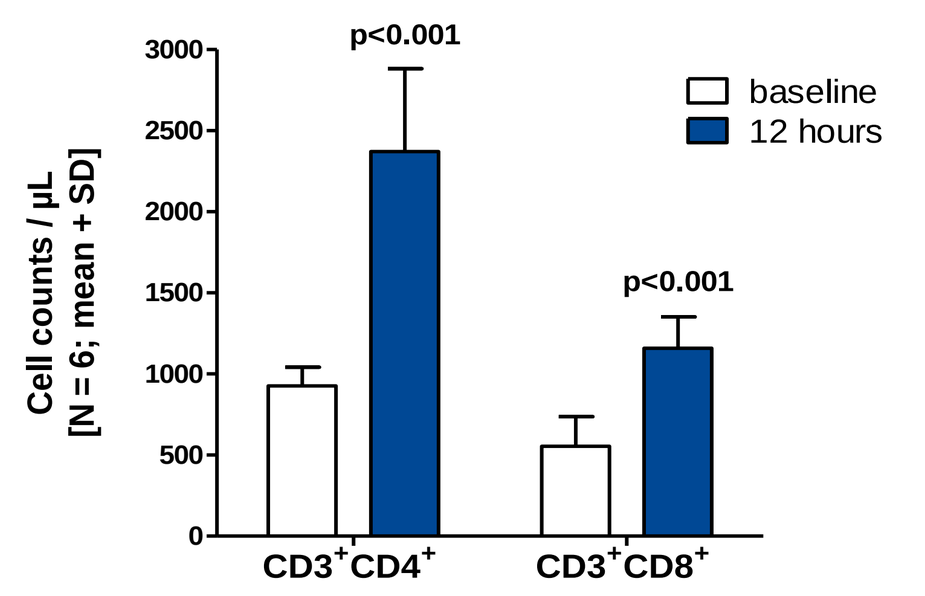

p<0.001
p<0.001
baseline
12 hours
Cell counts / µL [N = 6; mean + SD]
CD3+CD4+
CD3+CD8+

Figure 1. CD3+CD4+ and CD3+CD8+ T cells increase in peripheral blood 12 hours after NOX-A12 treatment (2mg/kg) in a Phase I clinical trial with healthy volunteers (unpublished data).

METHODS & RESULTS

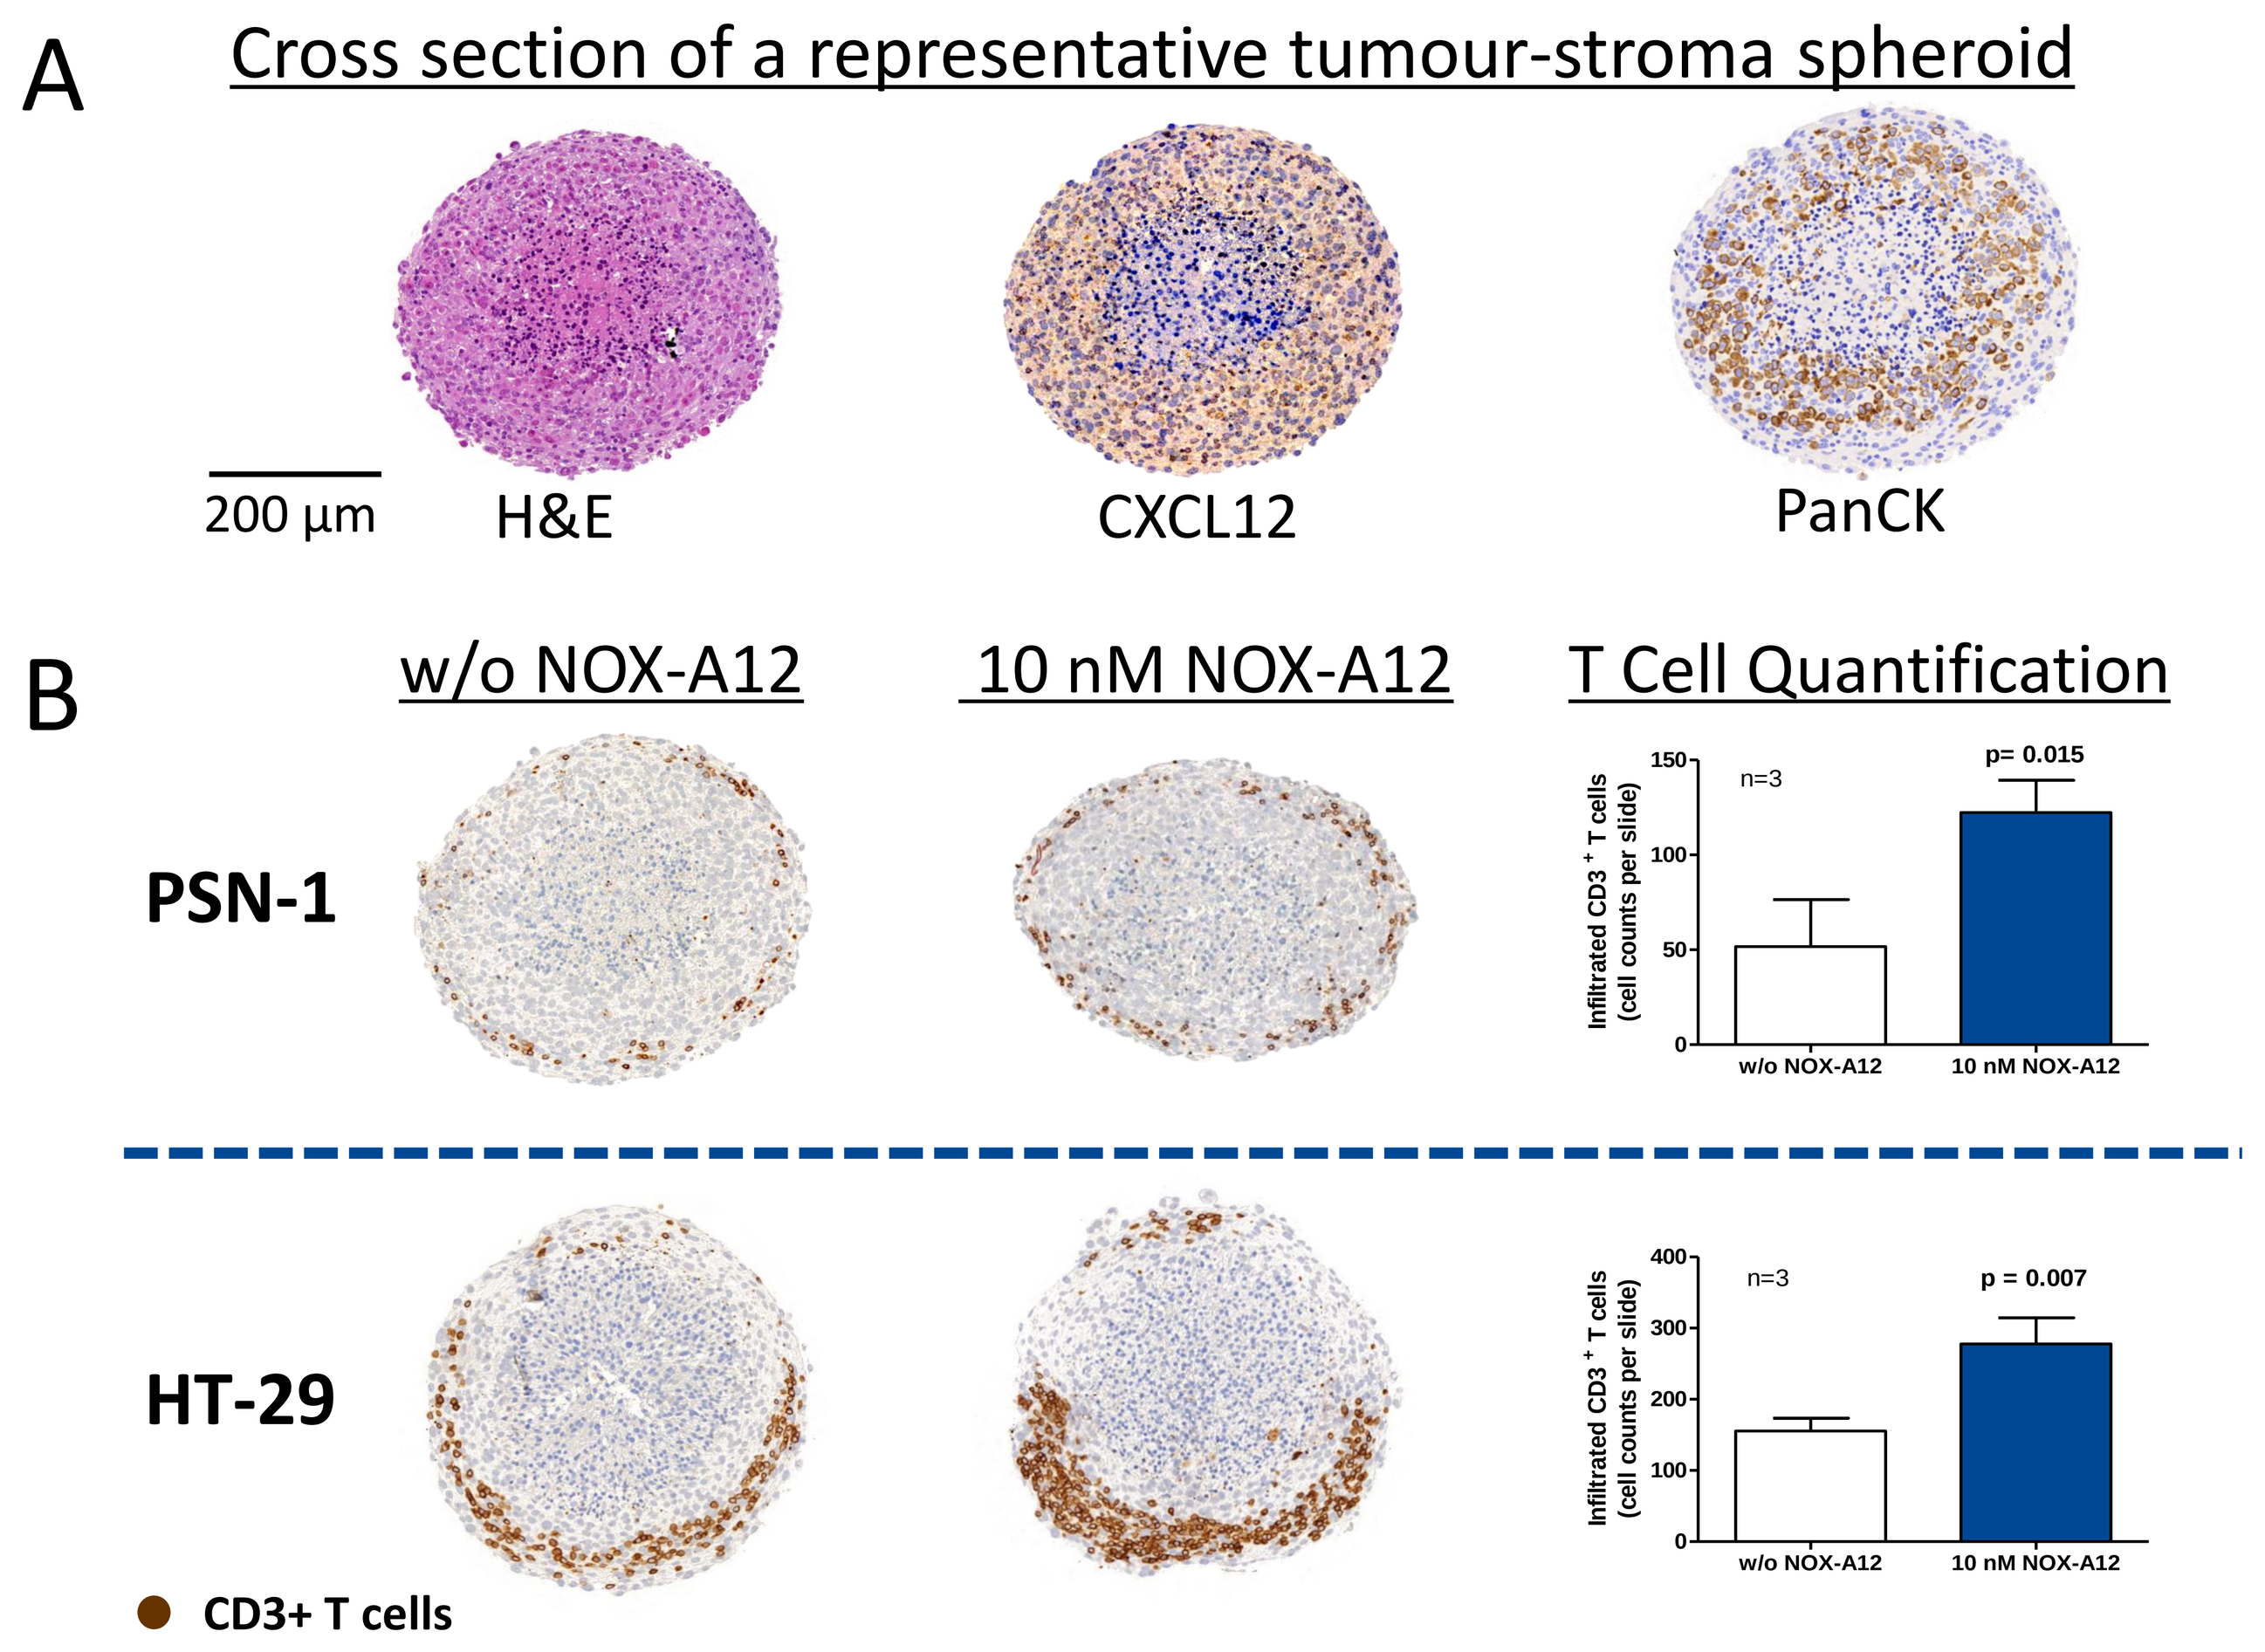

A
Cross section of a representative tumour-stroma spheroid
200 µm
H&E
CXCL12
PanCK
B
w/o NOX-A12
10 nM NOX-A12
T Cell Quantification
PSN-1
HT-29
n=3
p= 0.015
p = 0.007
Infiltrated CD3+ T cells (cell counts per slide)
w/o NOX-A12
10 nM NOX-A12
CD3+ T cells

Flow Cytometric Analysis of PBMC Infiltration

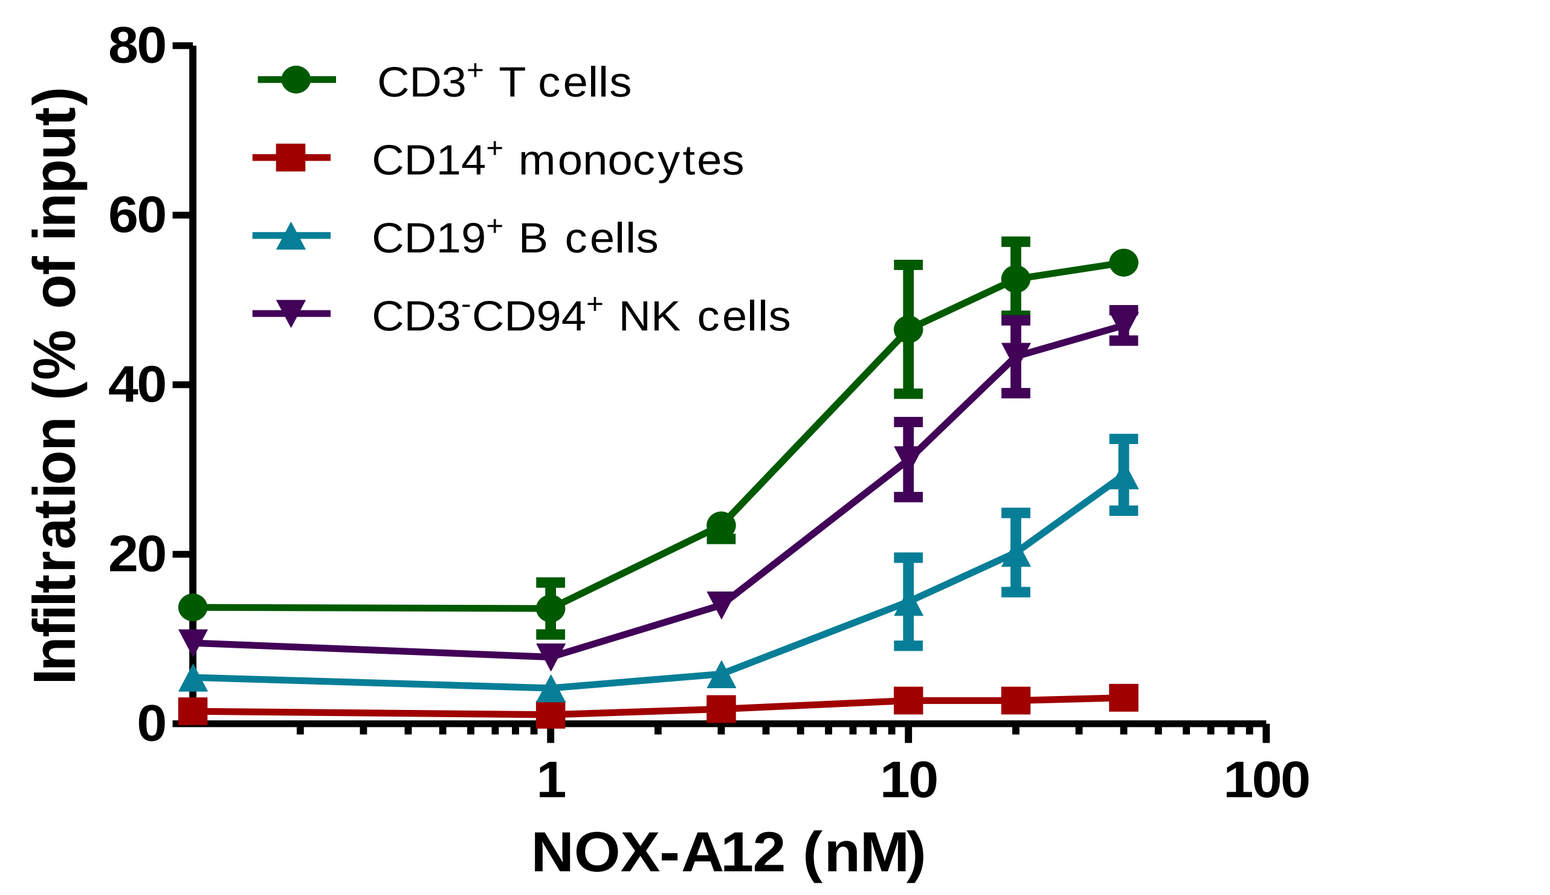

CD3+ T cells
CD14+ monocytes
CD19+ B cells
CD3-CD94+ NK cells
Infiltration (% of input)
NOX-A12 (nM)

Figure 3. Spheroids were generated by co-culturing of CXCL12-expressing MS-5 cells with human PSN-1 pancreatic cancer cells in ultra-low attachment plates. Primary human PBMCs were added to the spheroids in the presence or absence of various concentrations of NOX-A12. The next day, spheroids were washed and dissociated using Accumax (eBiosciences). Cells were incubated overnight to recover Accumax-labile cell surface molecules for detection by flow cytometry. Cell types were counted and normalized to the input cell count of each cell type.

Figure 2. CXCL12-expressing murine stromal MS-5 cells were co-cultured with human cancer cell lines (pancreatic PSN-1 (A,B), colorectal HT-29 (B)) in the ratio 5:1 in ultra-low attachment plates for 3 days. Isolated primary human T cells were added to the spheroids in the presence or absence of 10 nM NOX-A12. After overnight incubation spheroids were washed, formalin-fixed (2h) and paraffin-embedded. 2 µm sections were prepared for H&E- or immuno-staining. (A) PanCK: Pan cytokeratin = tumor cell marker (B) NOX-A12 increases the amount of T cells in both tumor-stroma spheroid models. Brown spots = CD3+ T cells.

RESULTS

A NOX-A12 Enhances T Cell Infiltration into Tumour-Stroma Spheroids

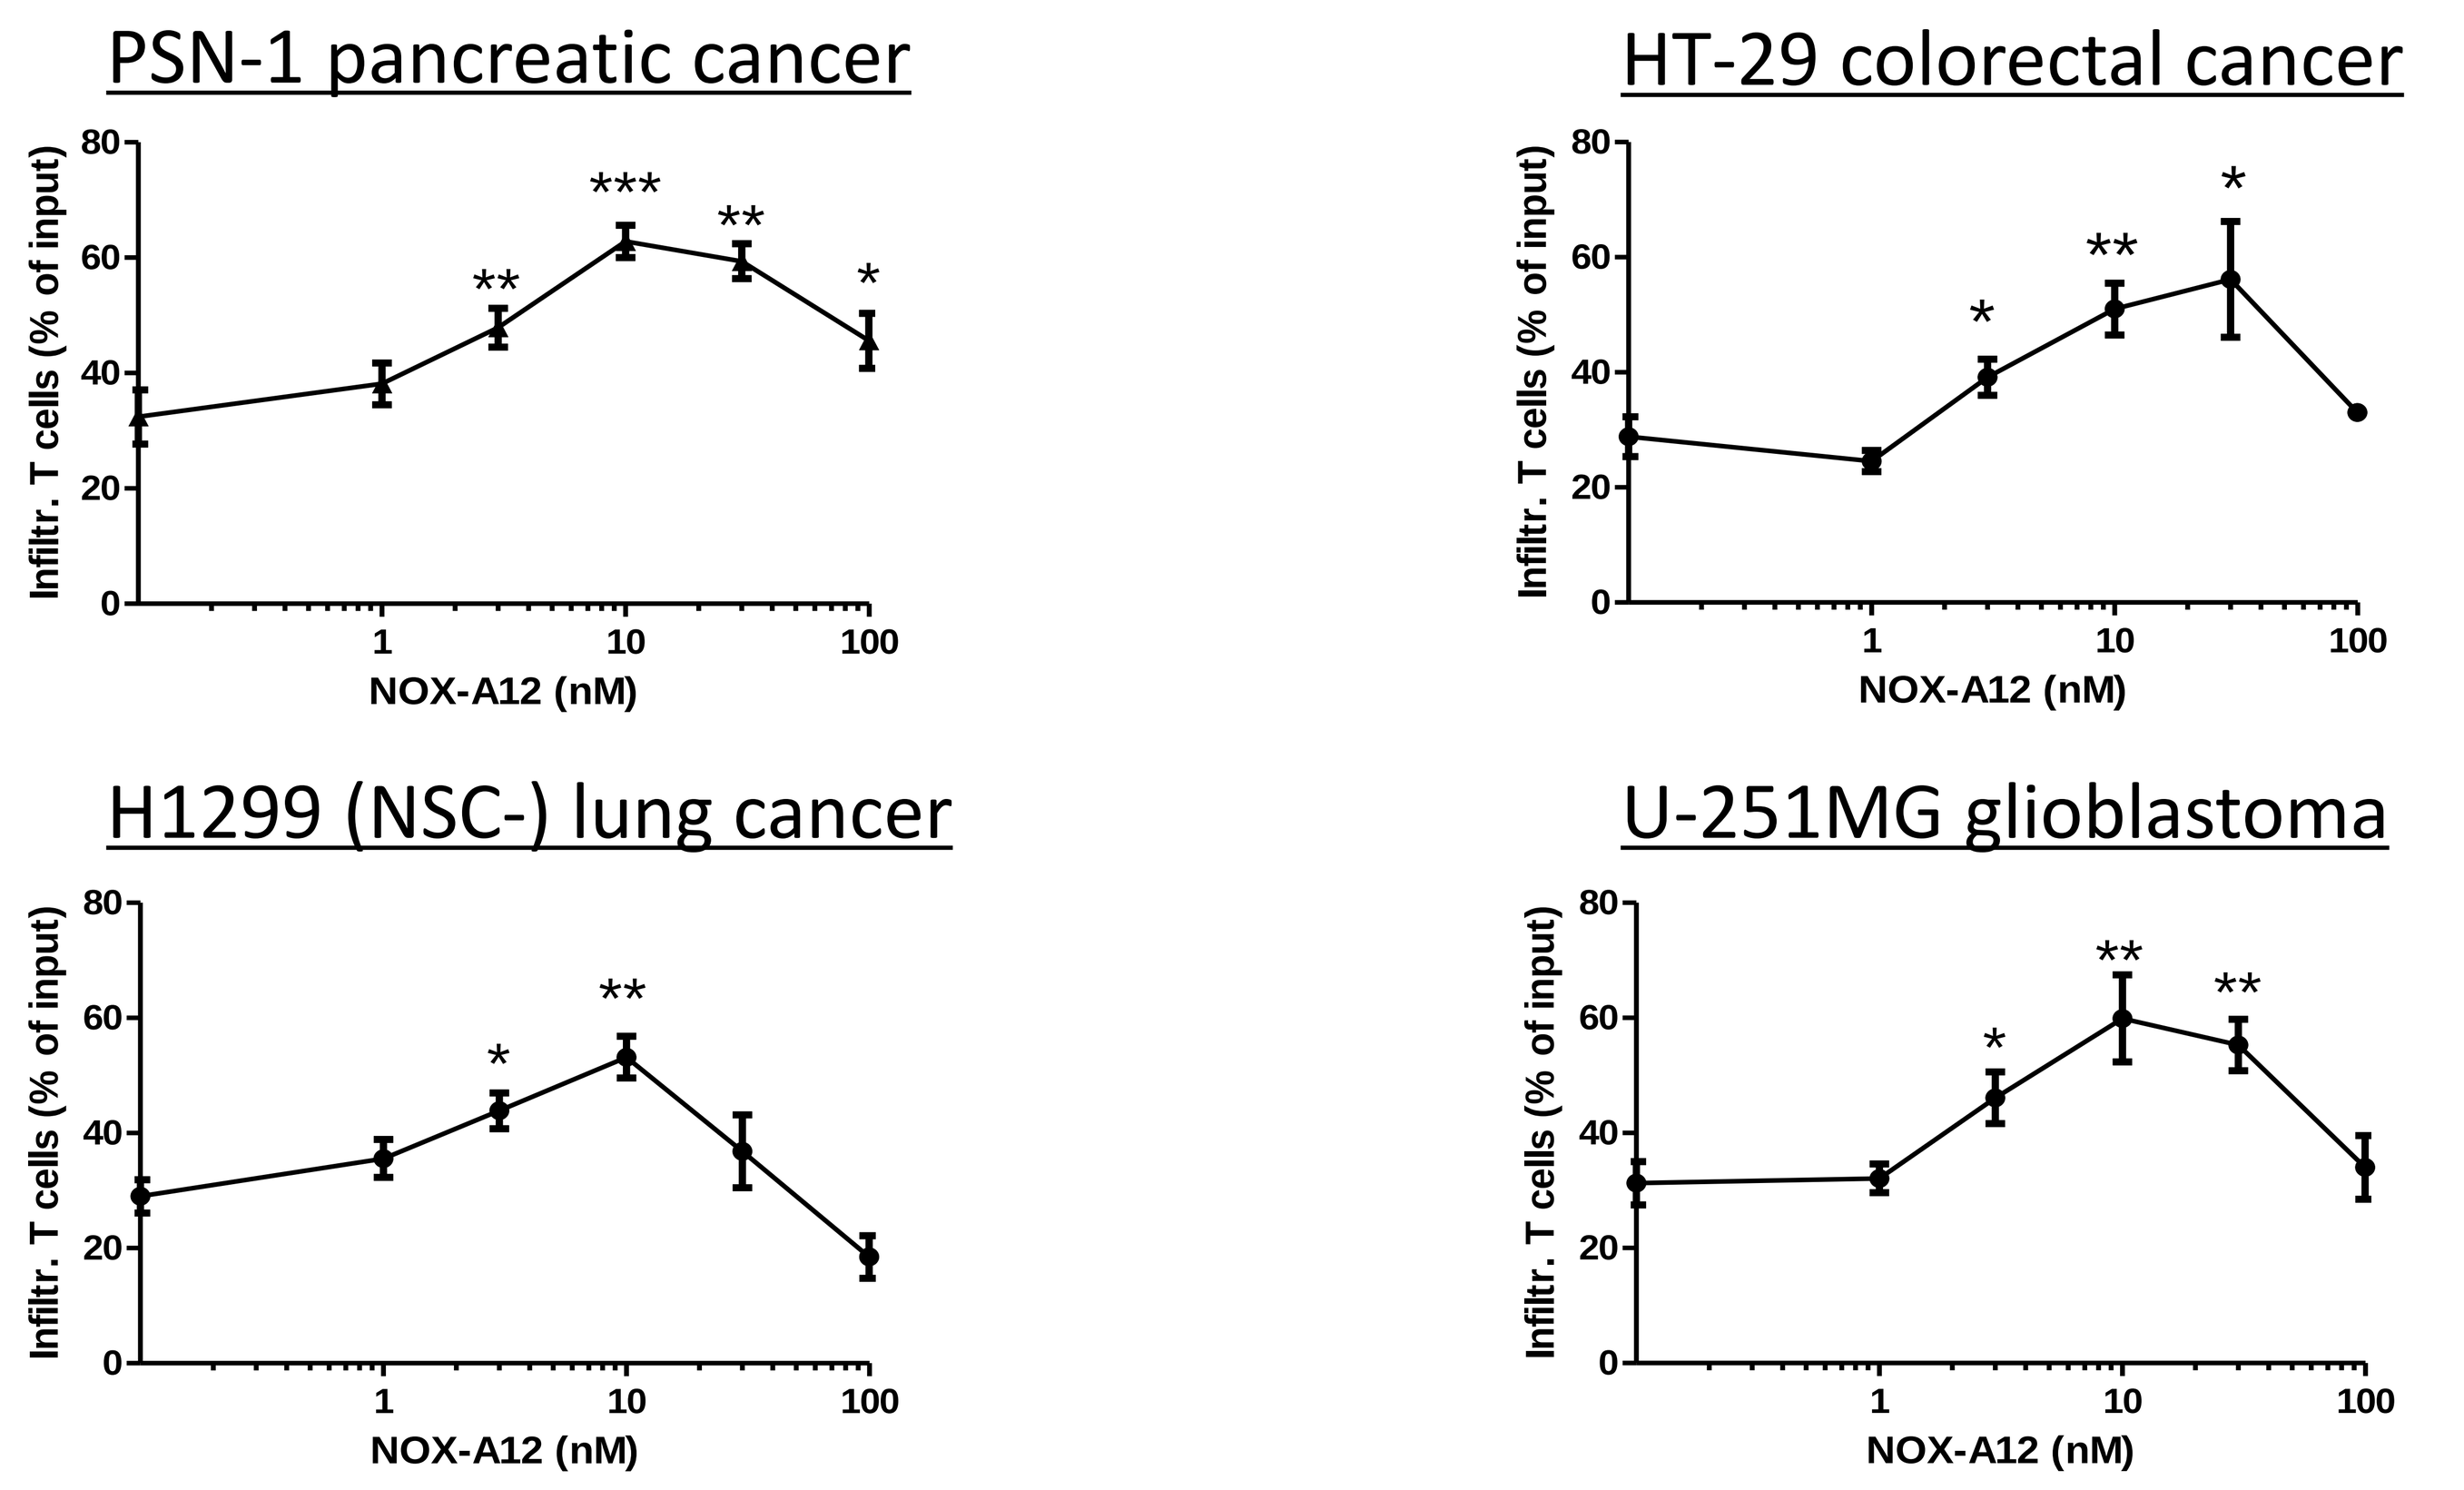

PSN-1 pancreatic cancer
HT-29 colorectal cancer
H1299 (NSC-) lung cancer
U-251MG glioblastoma
Infiltr. T cells (% of input)
NOX-A12 (nM)

B NOX-A12 Enhances NK Cell Infiltration into Tumour-Stroma Spheroids

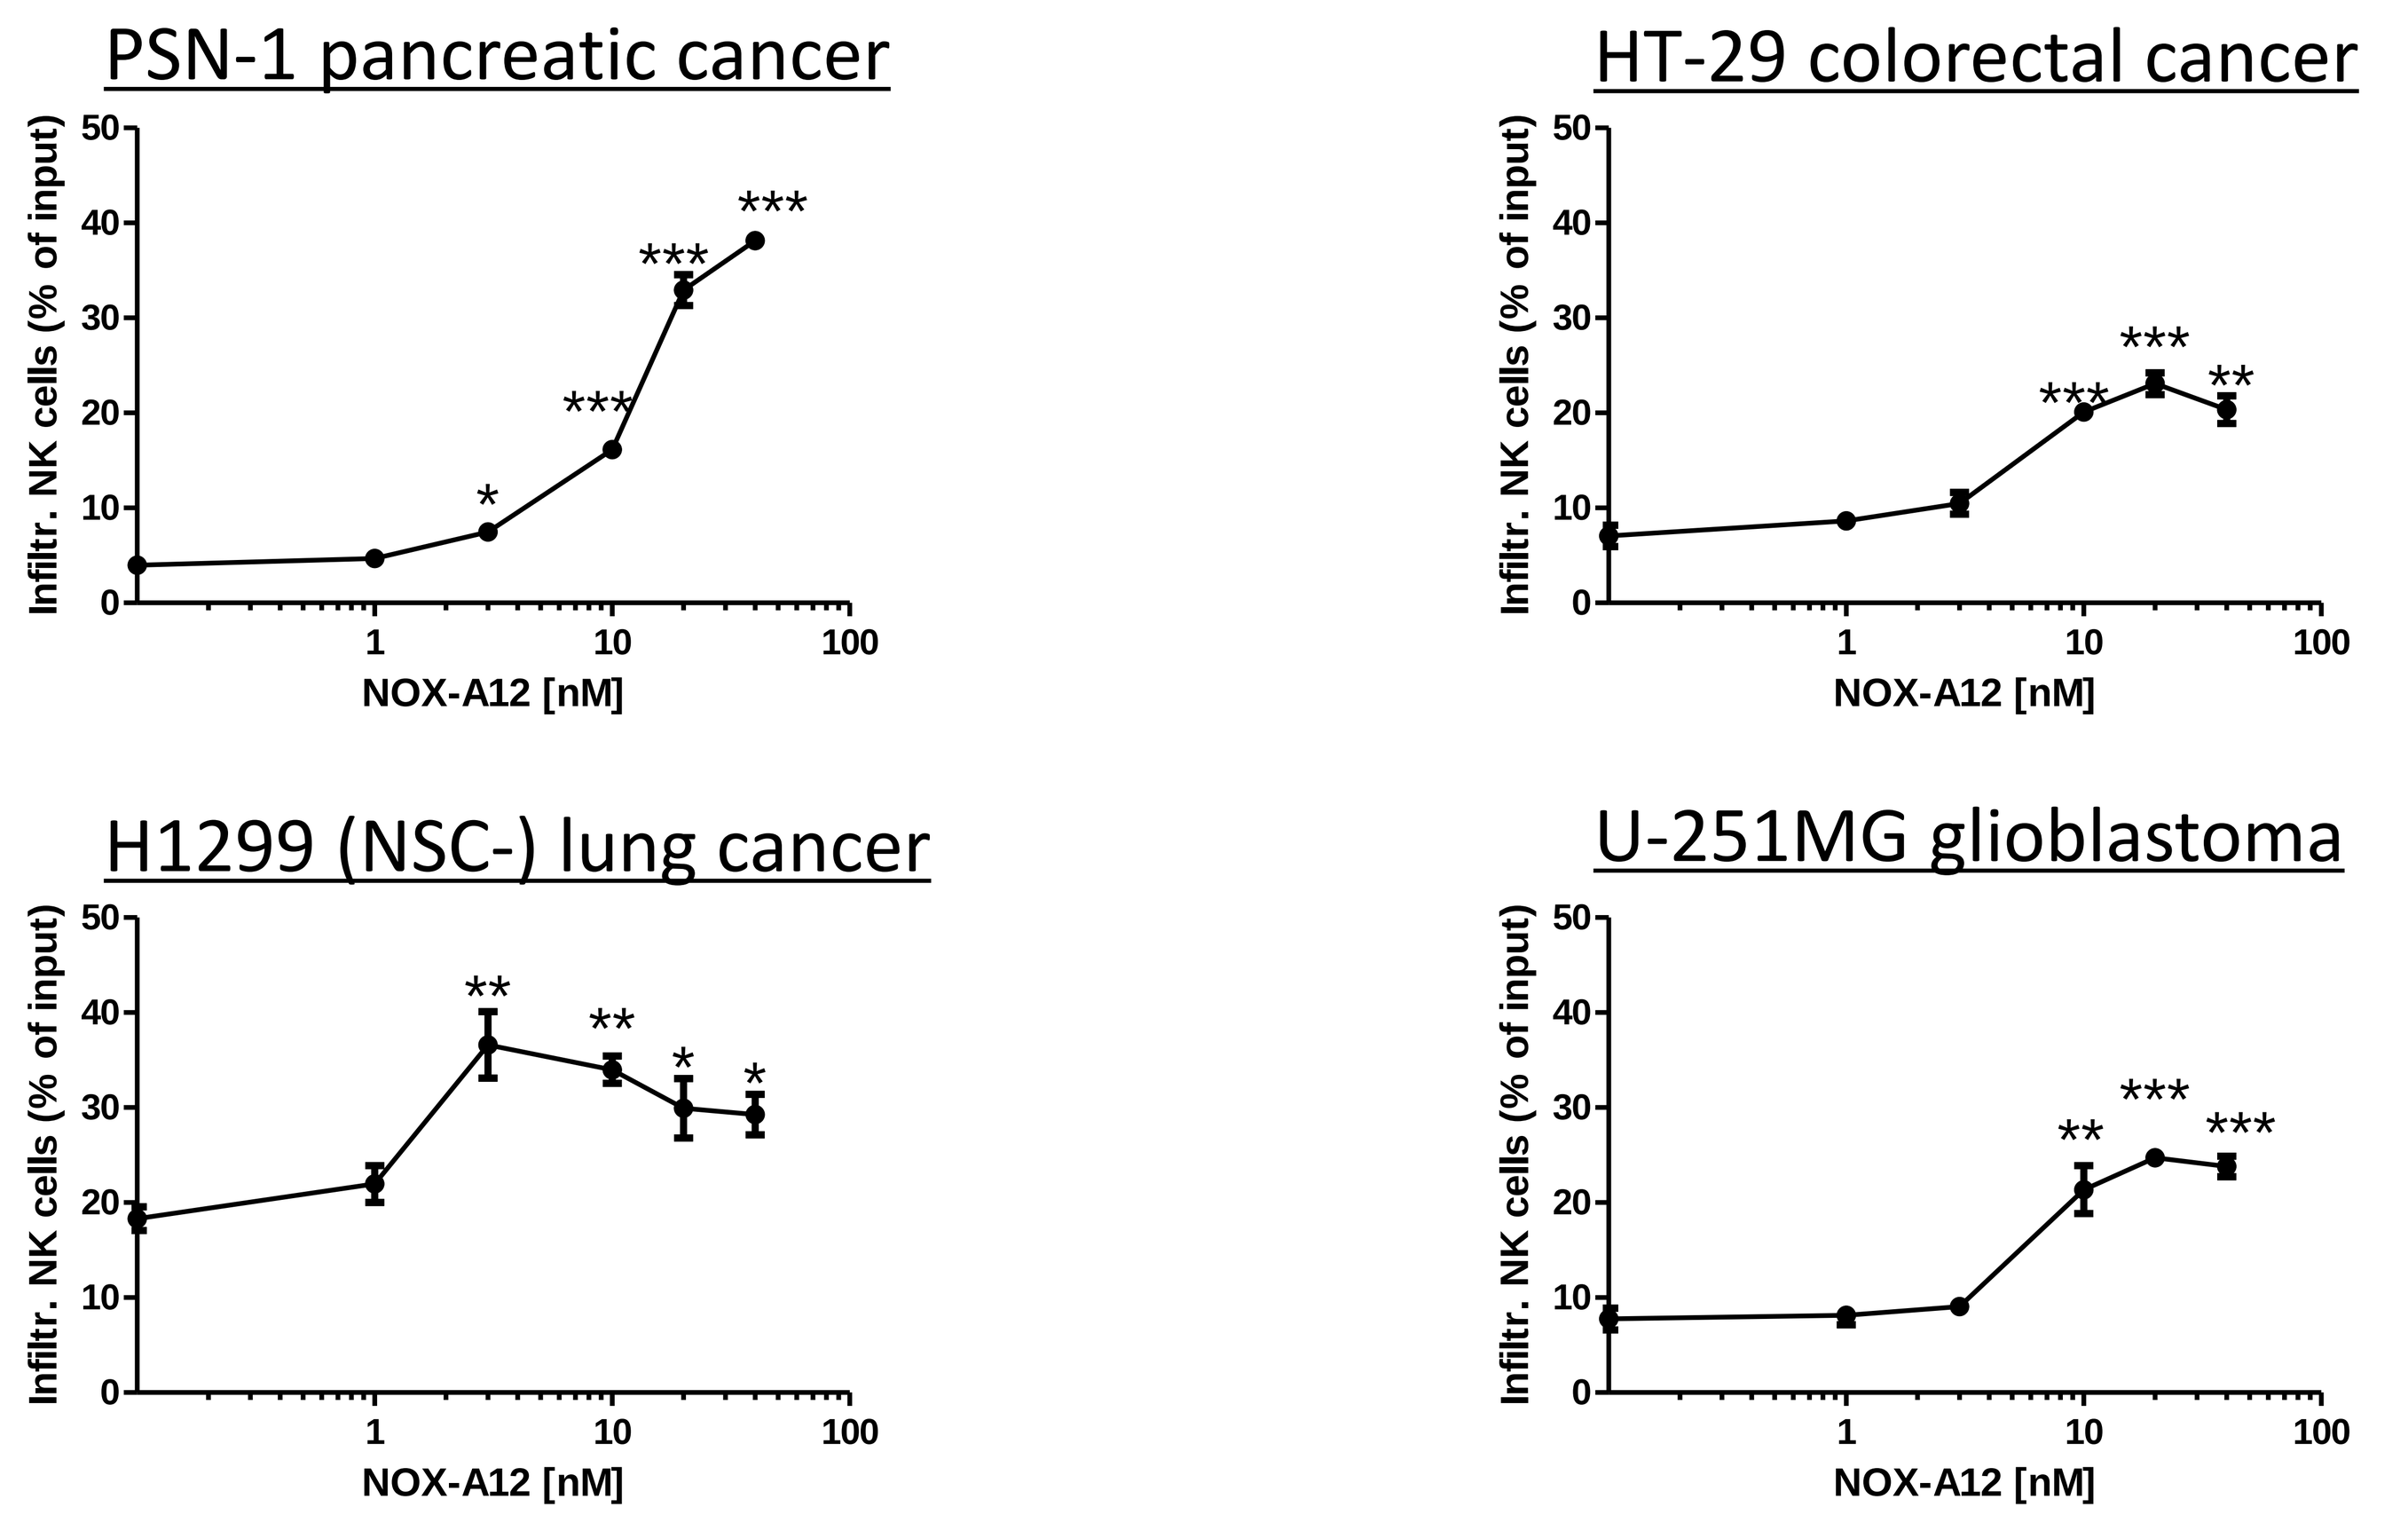

PSN-1 pancreatic cancer
HT-29 colorectal cancer
H1299 (NSC-) lung cancer
U-251MG glioblastoma
Infiltr. NK cells (% of input)
NOX-A12 [nM]

Figure 4. Spheroids of MS-5 cells with various tumor cell lines (PSN-1, HT-29, H1299 and U251MG) were generated as described in figure 2 and treated overnight with isolated primary T (A) or NK (B) cells from healthy donors in the presence of various concentrations of NOX-A12. The next day, spheroids were washed and dissociated for T or NK cell quantification by flow cytometry. *p<0.05; **p<0.01; ***p<0.001.

NOX-A12 Synergizes with T Cell-Based Checkpoint Inhibition (anti-PD-1 as Case Example)

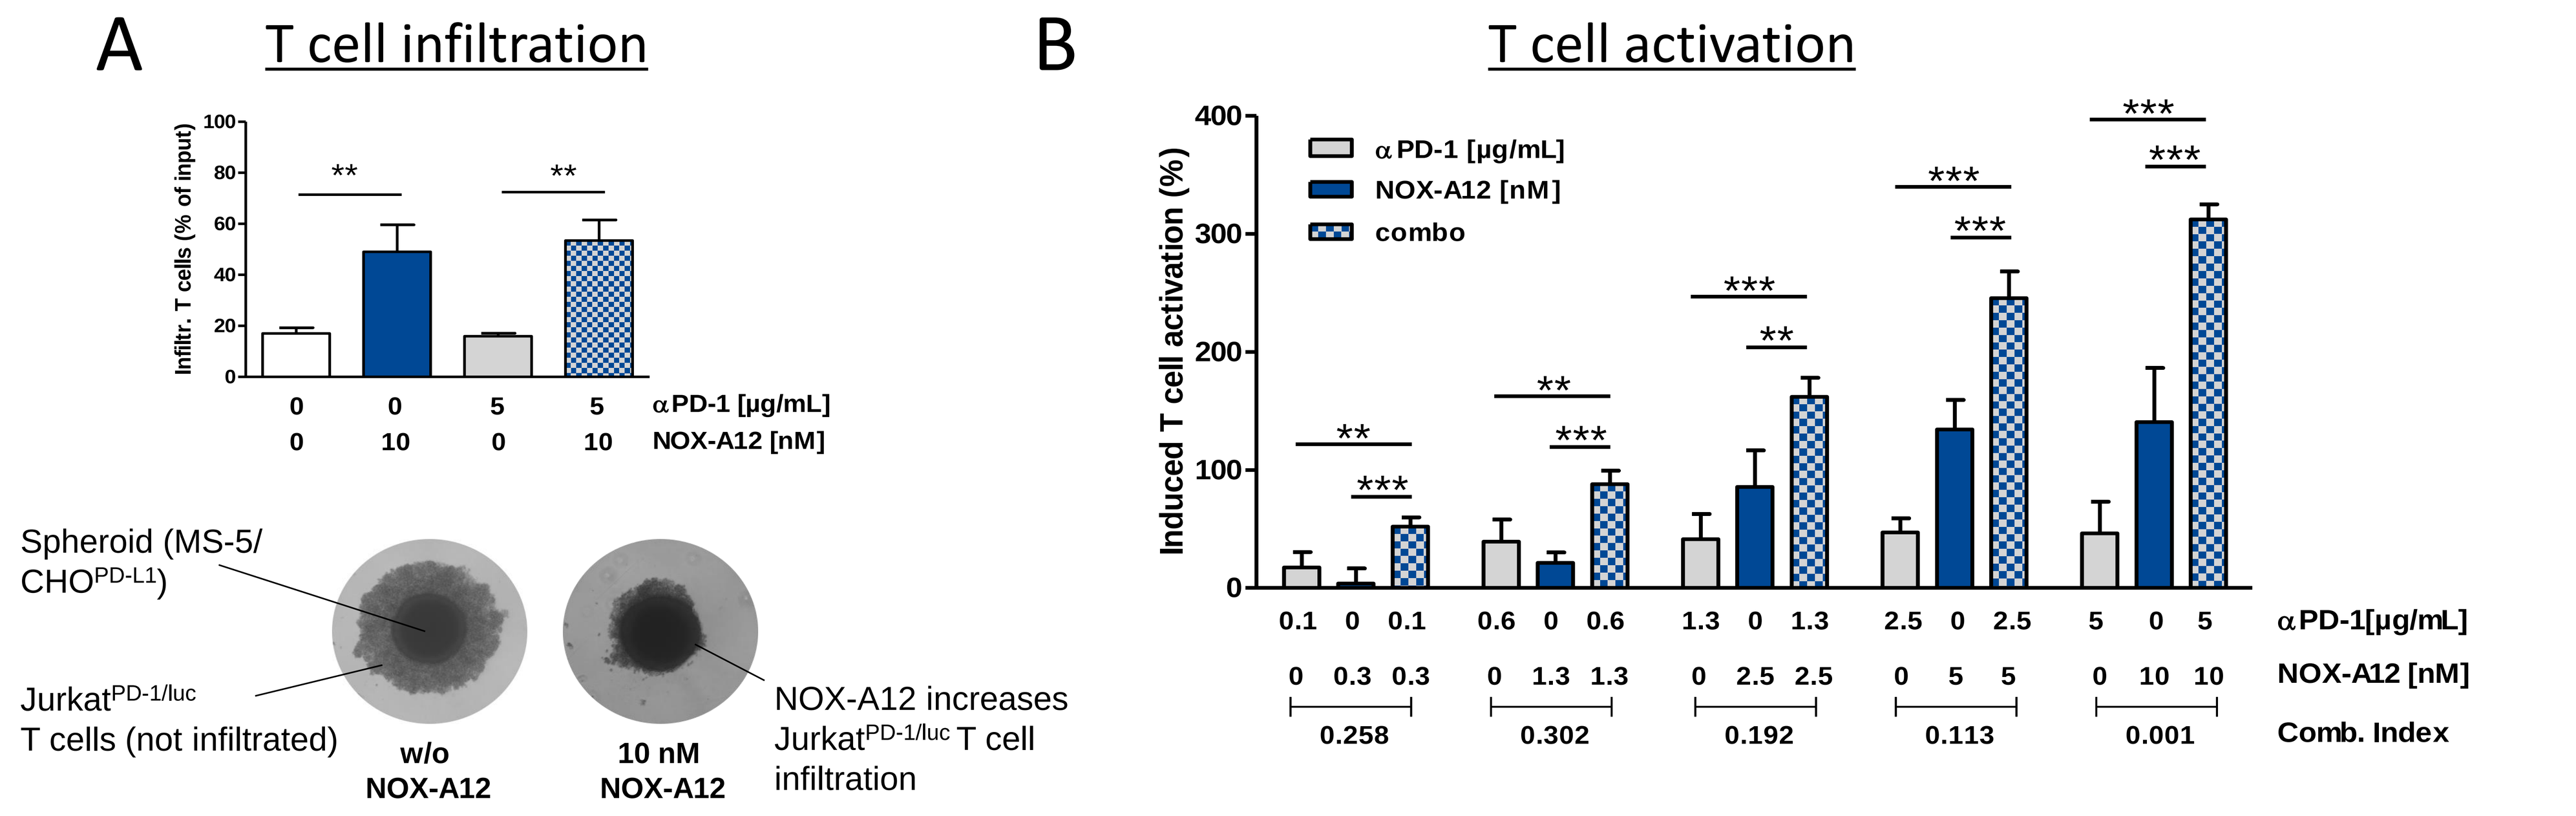

A
T cell infiltration
Infiltr. T cells (% of input)
αPD-1 [µg/mL]
NOX-A12 [nM]
Spheroid (MS-5/ CHOPD-L1)
JurkatPD-1/luc T cells (not infiltrated)
w/o NOX-A12
10 nM NOX-A12
NOX-A12 increases JurkatPD-1/luc T cell infiltration
B
T cell activation
αPD-1 [µg/mL]
NOX-A12 [nM]
combo
Induced T cell activation (%)
Comb. Index
0.258
0.302
0.192
0.113
0.001

Figure 5. The reporter-based PD-1/PD-L1 blockade bioassay from Promega was adapted to the 3D format: Jurkat$^{PD\text{-}1/luc}$ T cells were incubated with anti-PD-1 (clone PD1.3.1.3) and added to NOX-A12-treated tumor-stroma spheroids (CHO$^{PD\text{-}L1}$ + MS-5). The next day, T cell infiltration was quantified by flow cytometry and T cell activation by incubating the spheroids with BioGlo substrate (Promega). (A) NOX-A12, but not anti-PD-1, increases T cell infiltration which is also visible under the microscope. (B) T cell activation using anti-PD-1 is restricted by limited T cell - tumor cell contacts in the 3D spheroid structure. NOX-A12 dose-dependently increases T cell activation by facilitating T cell infiltration. NOX-A12 acts synergistically with anti-PD-1 (Comb. Index < 1).[10] *p<0.05; **p<0.01; ***p<0.001

NOX-A12 Synergizes with NK Cell-Mediated ADCC (Obinutuzumab as Case Example)

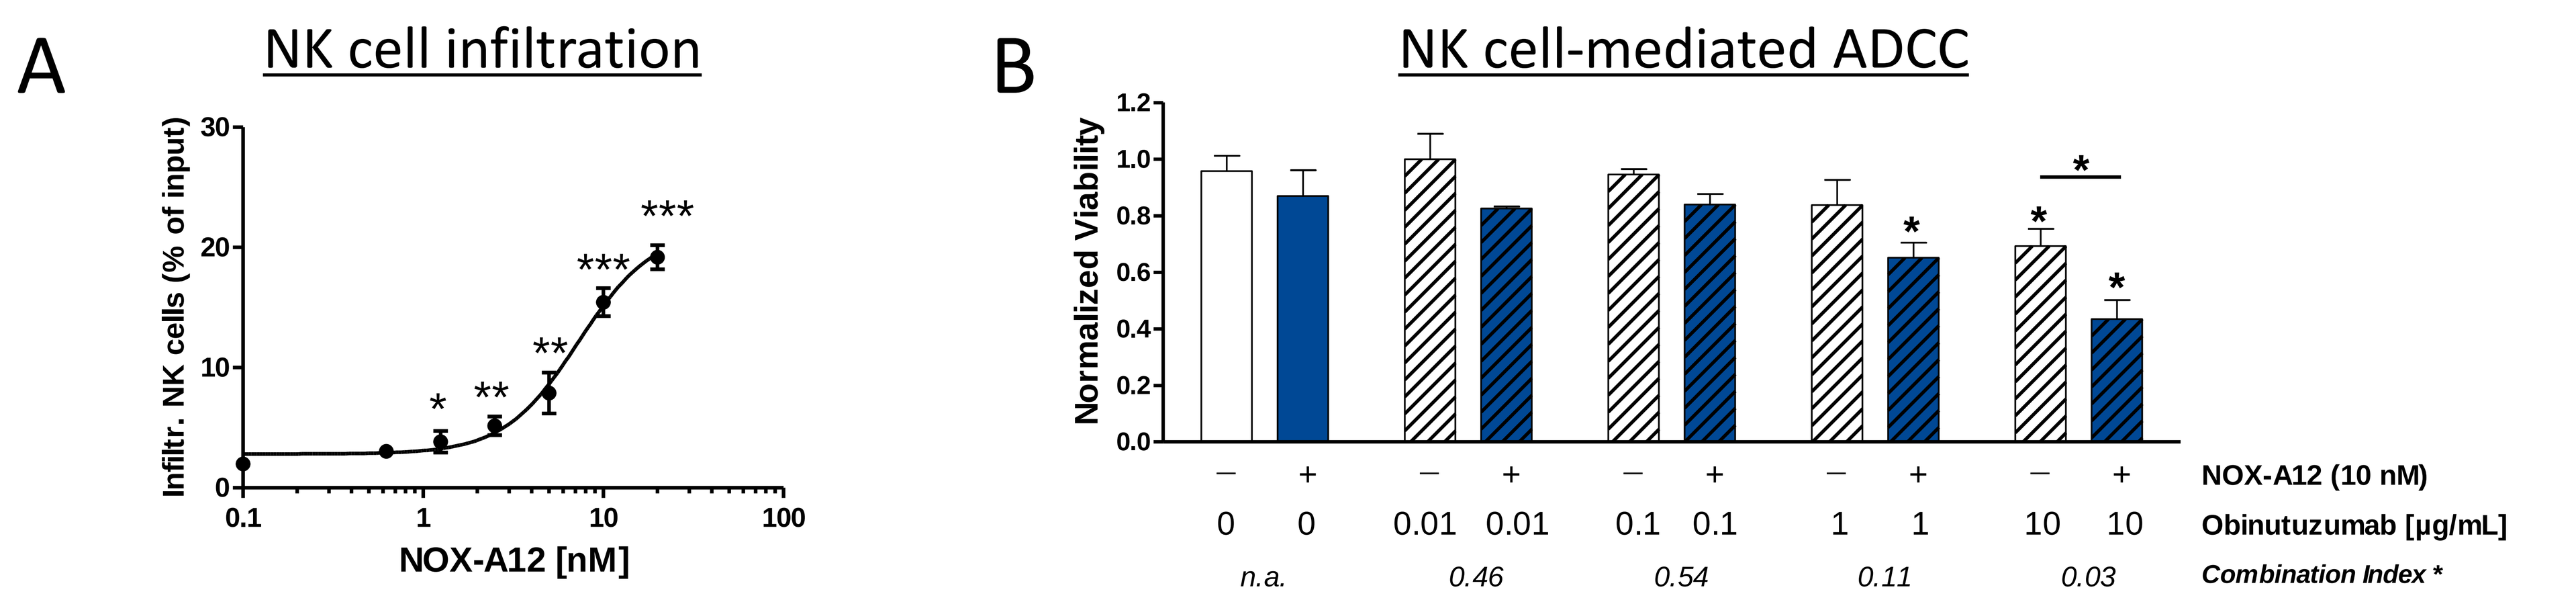

A
NK cell infiltration
Infiltr. NK cells (% of input)
NOX-A12 [nM]
B
NK cell-mediated ADCC
Normalized Viability
NOX-A12 (10 nM)
Obinutuzumab [µg/mL]
Combination Index *
n.a.
0.46
0.54
0.11
0.03

Figure 6. Spheroids of MS-5 cells with CFSE-stained Raji lymphoma cells were generated as described in figure 2 and treated overnight with primary human NK cells in the presence of various concentrations of NOX-A12. Spheroids were dissociated for NK cell quantification and determination of Raji cell viability by flow cytometry. NOX-A12 acts synergistically with obinutuzumab (Combination Index < 1).[10].

CONCLUSIONS & OUTLOOK

- We established tumour-stroma spheroids that mimic the complexity of the tumour microenvironment, in which the CXCL12 inhibitor NOX-A12 increases T and NK cell infiltration.
- By facilitating physical contact of both T and NK cells with tumour cells, NOX-A12 synergizes with T cell-based checkpoint inhibition and NK cell-mediated ADCC.
- This data suggests to combine NOX-A12 with T and NK cell-based cancer immunotherapy.

ESMO Congress 2016, Copenhagen , Denmark, 7-11 October 2016
Session Immunotherapy of cancer: Abstract #1083P
Location: Hall E, Sunday, Oct 9, 2016, 13 PM -14 PM

Conflict of Interest Disclosures : DZ, AK, DE, AV: NOXXON Pharma AG Employment.
"Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors."

References: (1): Joyce and Fearon, Science. 2015 Apr 3;348(6230):74-80; (2): Fearon, Cancer Immunol Res. 2014 Mar;2(3):187-93, (3): Hoellenriegel & Zboralski *et al.,* Blood. 2014 Feb 13;123(7):1032-9; (4): Roccaro *et al.,* Cell Rep. 2014 Oct 9;9(1):118-28; (5): Liu *et al.*, Neuro Oncol. 2014 Jan;16(1):21-8; (6): Vater *et al.,* Clin Pharmacol Ther. 2013 Jul;94(1):150-7; (7): Ludwig *et al.,* Blood. 2014 124:2111; (8): Steurer, *et al*. Blood. 2014 124:1996; (9): Chernikova *et al.* Mol Cancer Ther. 2013;12(11 Suppl):C291; (10): Chou, Cancer Res. 2010 Jan 15;70(2):440-6

Dirk Zboralski - Senior Scientist
NOXXON Pharma AG - In Vitro Pharmacology
Email: zbo@gmx.net